Abstract Number
818

A logic-gated CAR T for post-transplant acute myeloid leukemia that distinguishes donor and recipient hematopoiesis

Diane Manry[1], Breanna L. Luna[1], Aaron Winters[1], Aaron D. Flynn[1], Tanveer Gill[1], Michele McElvain[1], Alaia Sima[1], Jason Wang[1], Alexandre Zampieri[1], Jingli Zhang[1], Sara Martire[1], Breanna DiAndreth[1], Chawita Netirojjanakul[1], Talar Tokatlian[1], Alexander Kamb[1], John S. Welch[1]

[1]A2 Biotherapeutics, Agoura Hills, California, USA

ABSTRACT

Background: AML relapse after a stem cell transplant is associated with dismal outcomes and no current FDA-approved therapeutic options. Cell therapy for AML has been challenging due to a lack of antigens distinguishing malignant from normal hematopoiesis, the potential for antigen escape due to intratumor antigen heterogeneity, and poor T cell function in lymphocytes from AML patients. To overcome these problems, we focus on opportunities in patients who have received HLA-A*02 mismatched transplants.

We have developed a Tmod HLA-A*02 NOT gate (blocker) that prevents CAR T killing of HLA-A*02 positive cells. In patients who are HLA-A*02 negative and receive an HLA-A*02 positive stem cell transplant, the blocker distinguishes tumor (HLA-A*02 negative) and normal hematopoiesis (HLA-A*02 positive). A similar Tmod NOT gate has been safe in an ongoing solid tumor clinical trial (NCT05736731)(Grierson SITC 2024). This is paired with a dual CD33/CLL-1 activator to prevent antigen escape and can use donor-derived HLA-A*02 positive T cells to mitigate AML patient T cell dysfunction. Additional blockers targeting HLA-A*03, etc. can expand therapeutic opportunity to patients who have received other HLA mismatch transplants.

Methods: CAR constructs were designed and then screened in Jurkat and primary T cell assays. EC50 and IC50 results were compared to monospecific CAR constructs using benchmark activators based on scFvs from M26 (CLL-1) and lintuzumab (CD33) using wild-type A*02-negative and syngeneic A*02-positive engineered AML cell lines.

Results: 111 tandem CD33/CLL-1 activator CAR constructs were evaluated in Jurkat cells and 23 in conjunction with HLA-A*02 blocker. Six tandem constructs were prioritized for evaluation in primary T cells, identifying one (20I21-1A4 mBAAsh) with IC50 potency similar to monospecific CAR constructs, and selectivity observed between HLA-A*02 positive vs. HLA-A*02 negative cells. Selective protection also was observed in HLA-A*02 positive primary hematopoietic stem cells compared with CAR constructs without the blocker.

Conclusions: We have developed a cell therapy strategy to leverage HLA mismatched stem cell transplants using a Tmod HLA-A*02 blocker. This approach could be applied to patients who have relapsed after a mismatched transplant or integrated into a stem cell transplant to prevent relapse. Pre-clinical data of this design demonstrates selectivity between HLA-A*02 positive and HLA-A*02 negative cells, and dual antigen activity addresses antigen escape. Future studies will interrogate the impact of membrane-tethered cytokines to augment potency and xenografts studies to further define activity.

Figure 1: Schematic for pairing Tmod with an allogeneic stem cell transplant to treat AML

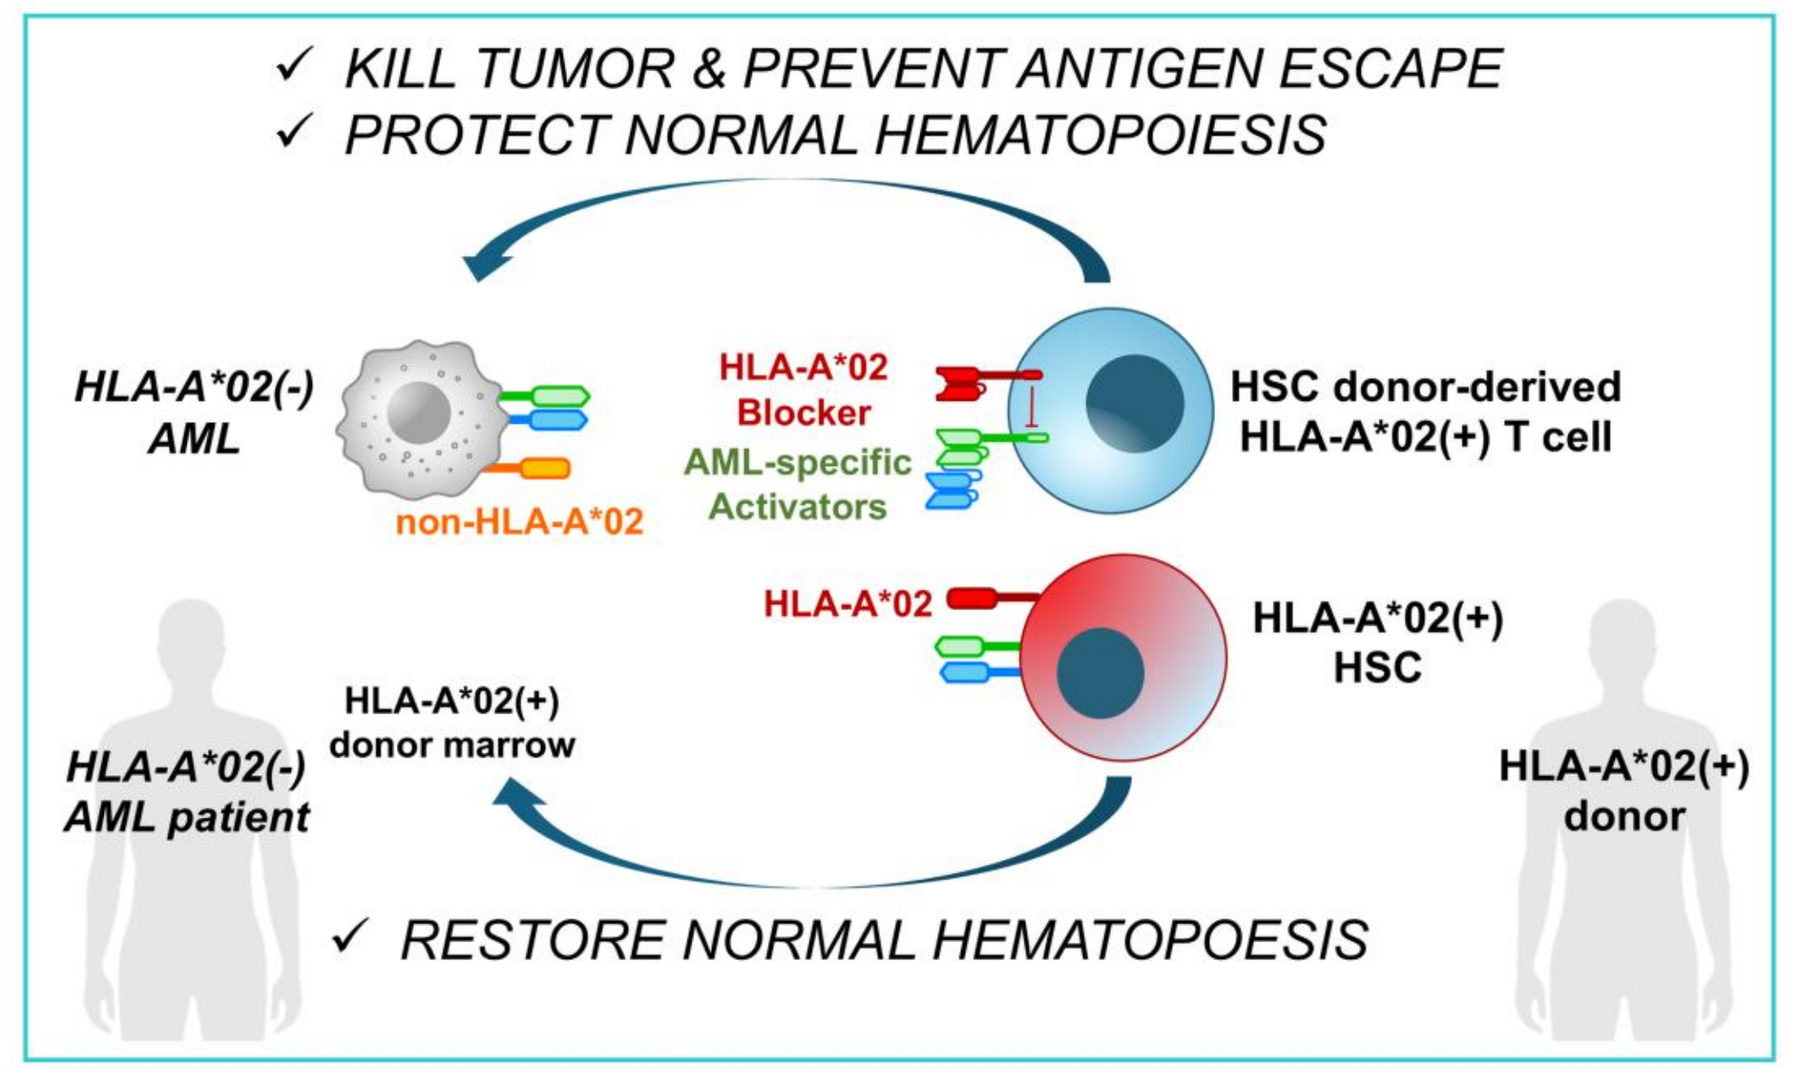
✓ KILL TUMOR & PREVENT ANTIGEN ESCAPE
✓ PROTECT NORMAL HEMATOPOIESIS
HLA-A*02(-) AML
non-HLA-A*02
HLA-A*02 Blocker
AML-specific Activators
HSC donor-derived HLA-A*02(+) T cell
HLA-A*02
HLA-A*02(+) HSC
HLA-A*02(+) donor marrow
HLA-A*02(-) AML patient
HLA-A*02(+) donor
✓ RESTORE NORMAL HEMATOPOESIS

Figure 1: A method of treating patients with AML by combining a stem cell transplant with a cell therapy product containing a tandem CD33-CLL-1 activator and a Tmod HLA-A*02 NOT gate (blocker). In this scenario, an HLA-A*02(-) patient receives a stem cell transplant and Tmod T cells from the same HLA-A*02+ donor. Tmod T cells recognize and kill AML cells expressing CD33 and/or CLL-1, while protecting the HLA-A*02+ transplanted stem cells and donor-derived hematopoiesis via the HLA-A*02 blocker.

CD33 AND CLL-1 ARE HIGHLY EXPRESSED ON AML CELLS, BUT ALSO ON CRITICAL NORMAL CELL TYPES

Figure 2: CD33 and CLL-1 expression on AML and normal cell types

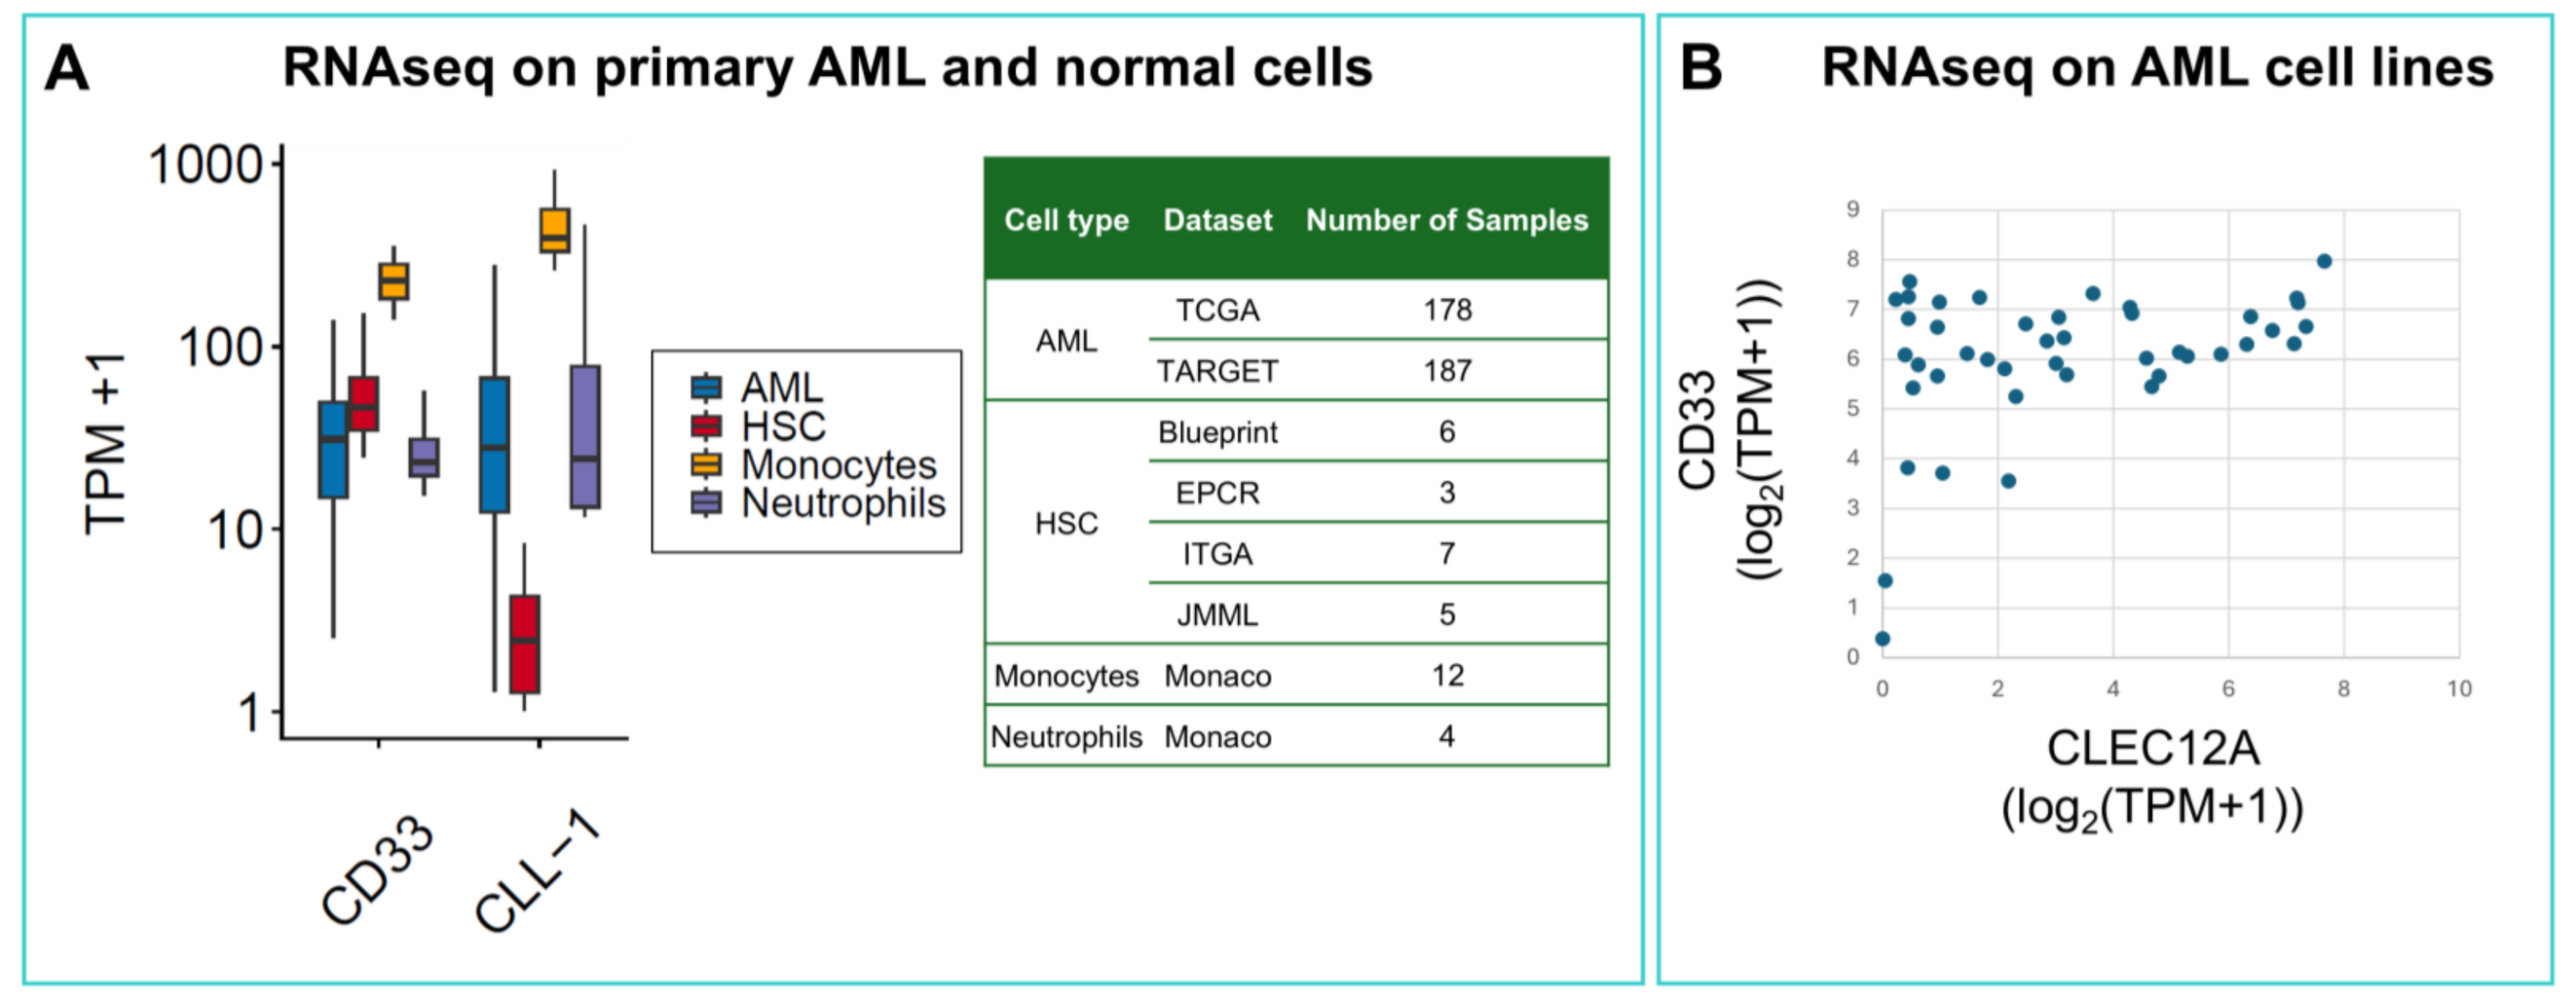
A
RNAseq on primary AML and normal cells
TPM +1
AML
HSC
Monocytes
Neutrophils
CD33
CLL-1
B
RNAseq on AML cell lines
CD33 (log2(TPM+1))
CLEC12A (log2(TPM+1))

Cell type	Dataset	Number of Samples
AML	TCGA	178
	TARGET	187
HSC	Blueprint	6
	EPCR	3
	ITGA	7
	JMML	5
Monocytes	Monaco	12
Neutrophils	Monaco	4

Figure 2: (A) CLL-1 and CD33 are both highly expressed on AML, but are also expressed on critical normal tissues, such as monocytes, neutrophils, and hematopoietic stem cells [1-7]. (B) CD33 and CLL-1 (CLEC12A) expression are uncorrelated in AML cell lines, supporting the utility of a dual-targeting approach to prevent antigen escape [8].

A TANDEM CLL-1-CD33 CAR DEMONSTRATES ACTIVATION TO BOTH ANTIGENS

Figure 3: Monospecific and tandem CARs targeting CD33 and/or CLL-1 exhibit dose-dependent activation to antigen(s)

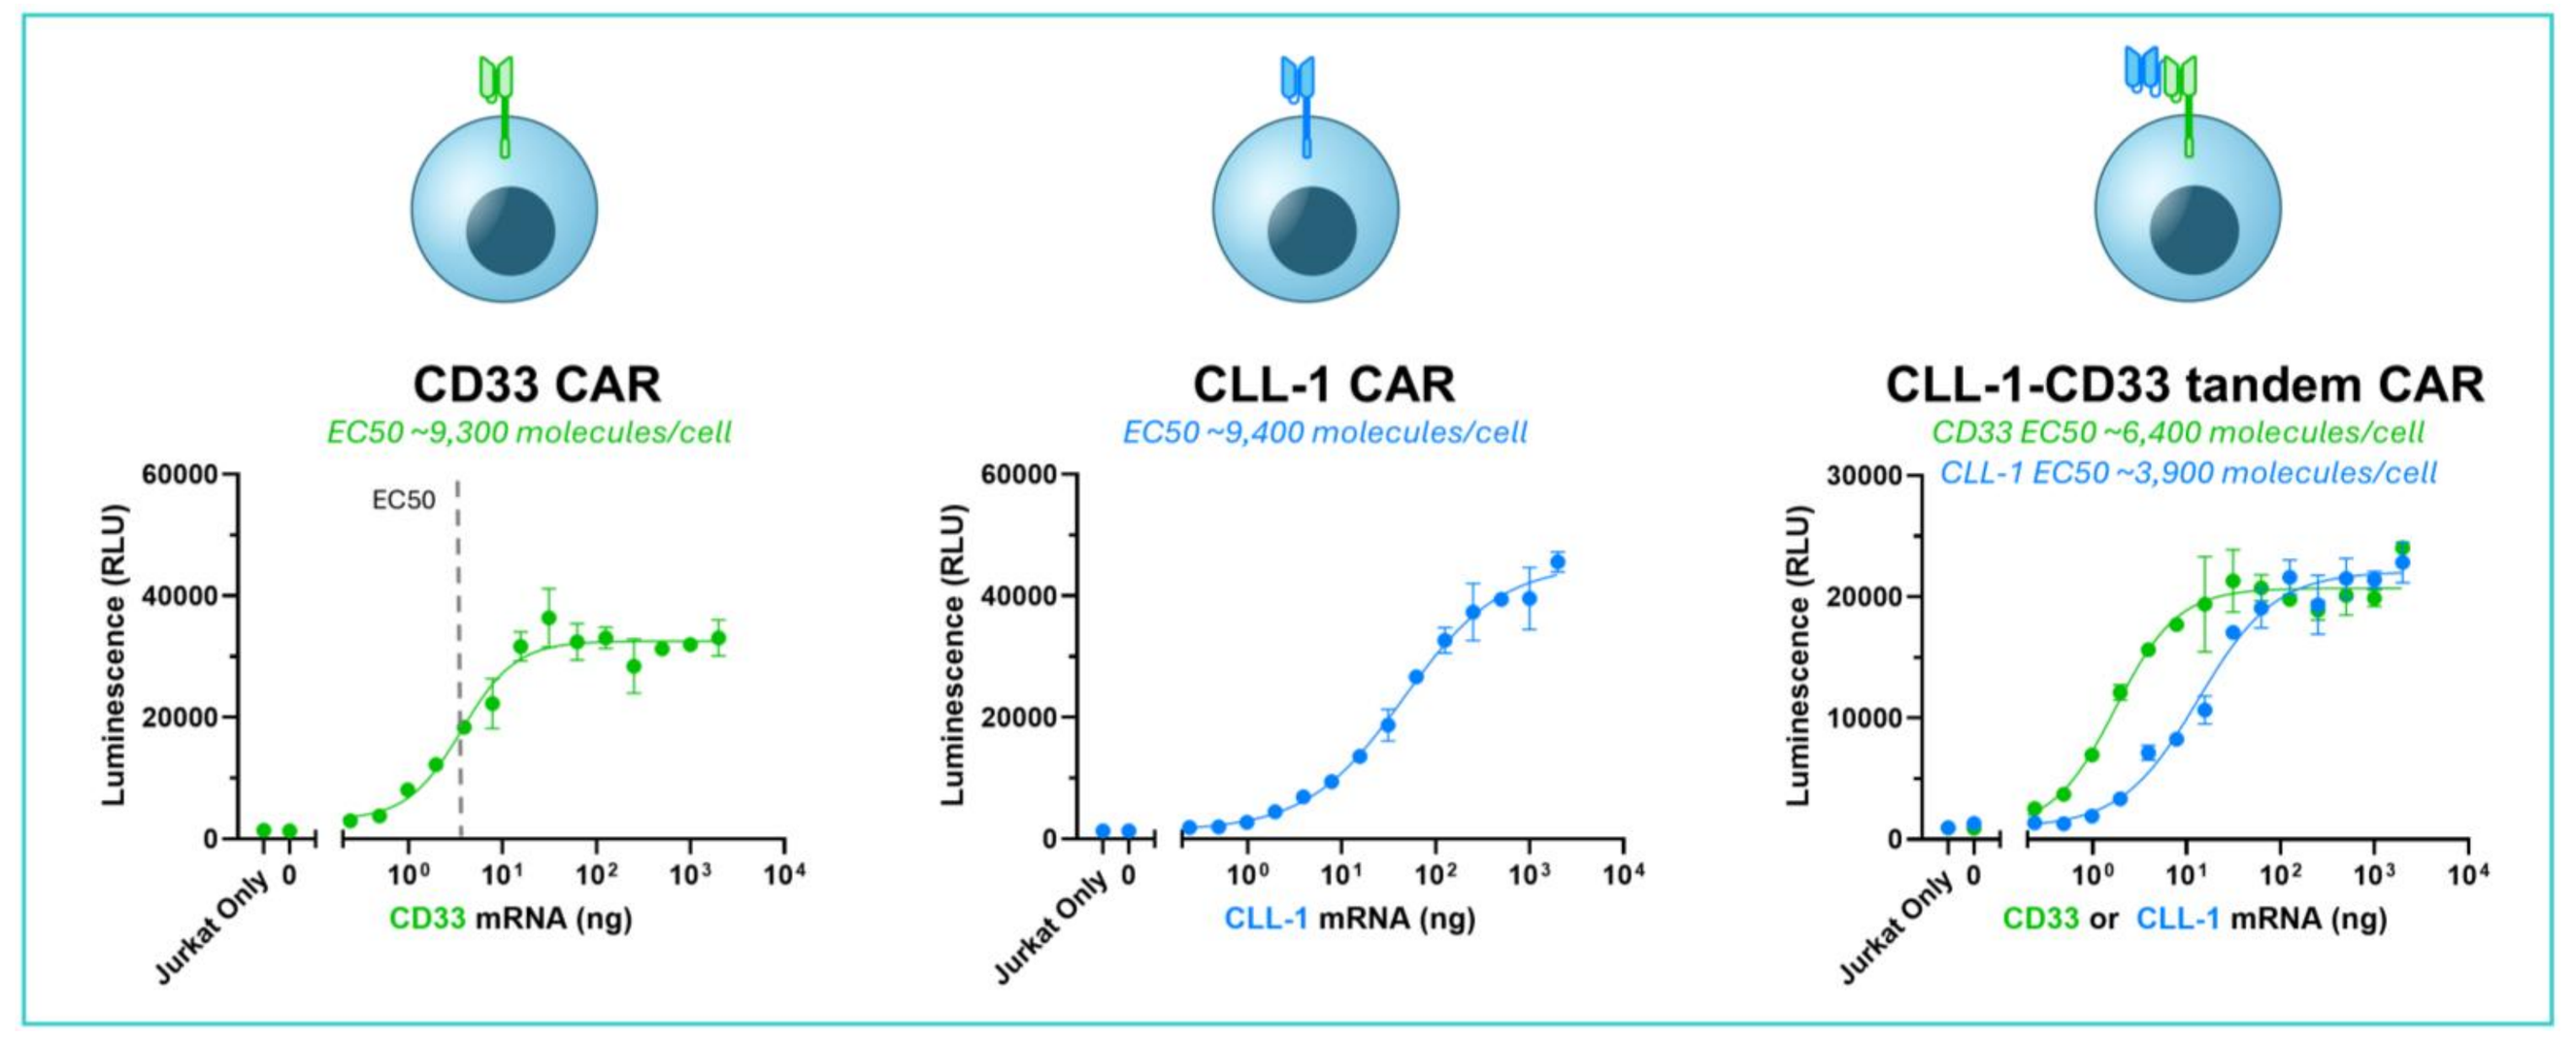
CD33 CAR
EC50 ~9,300 molecules/cell
CLL-1 CAR
EC50 ~9,400 molecules/cell
CLL-1-CD33 tandem CAR
CD33 EC50 ~6,400 molecules/cell
CLL-1 EC50 ~3,900 molecules/cell
Luminescence (RLU)
Jurkat Only
CD33 mRNA (ng)
CLL-1 mRNA (ng)
CD33 or CLL-1 mRNA (ng)

Figure 3: Monospecific CD33 (left) or CLL-1 (center) CARs as well as a tandem CLL-1-CD33 CAR (right) demonstrate dose-dependent activation to their target antigen(s) in Jurkat cells. Jurkat cells were transiently transfected with CAR constructs and co-cultured with K562 target cells transfected with titrated CD33 or CLL-1 mRNA. NFAT activation was measured via luciferase reporter after 6-hour co-culture of Jurkat and K562 cells.

AML-TARGETING CARS CAN BE INHIBITED BY MHC CLASS I-TARGETING BLOCKERS

Tmod is a dual-receptor NOT gate system comprised of an activator receptor (CAR or TCR) and a LILRB1-derived blocker receptor that inhibits activation when engaged with its target antigen [9]. A blocker targeting HLA-A*02 has demonstrated safety in an ongoing clinical trial (NCT05736731)(Grierson SITC 2024). This blocker, along with another blocker targeting HLA-A*03, both demonstrated robust blocking of CD33, CLL-1, and CLL-1-CD33 CAR activation in Jurkat cells (**Figure 4**).

Figure 4: Monospecific CARs targeting CD33 or CLL-1 as well as tandem CARs targeting both antigens can be blocked by HLA-A*02 or HLA-A*03-directed blockers

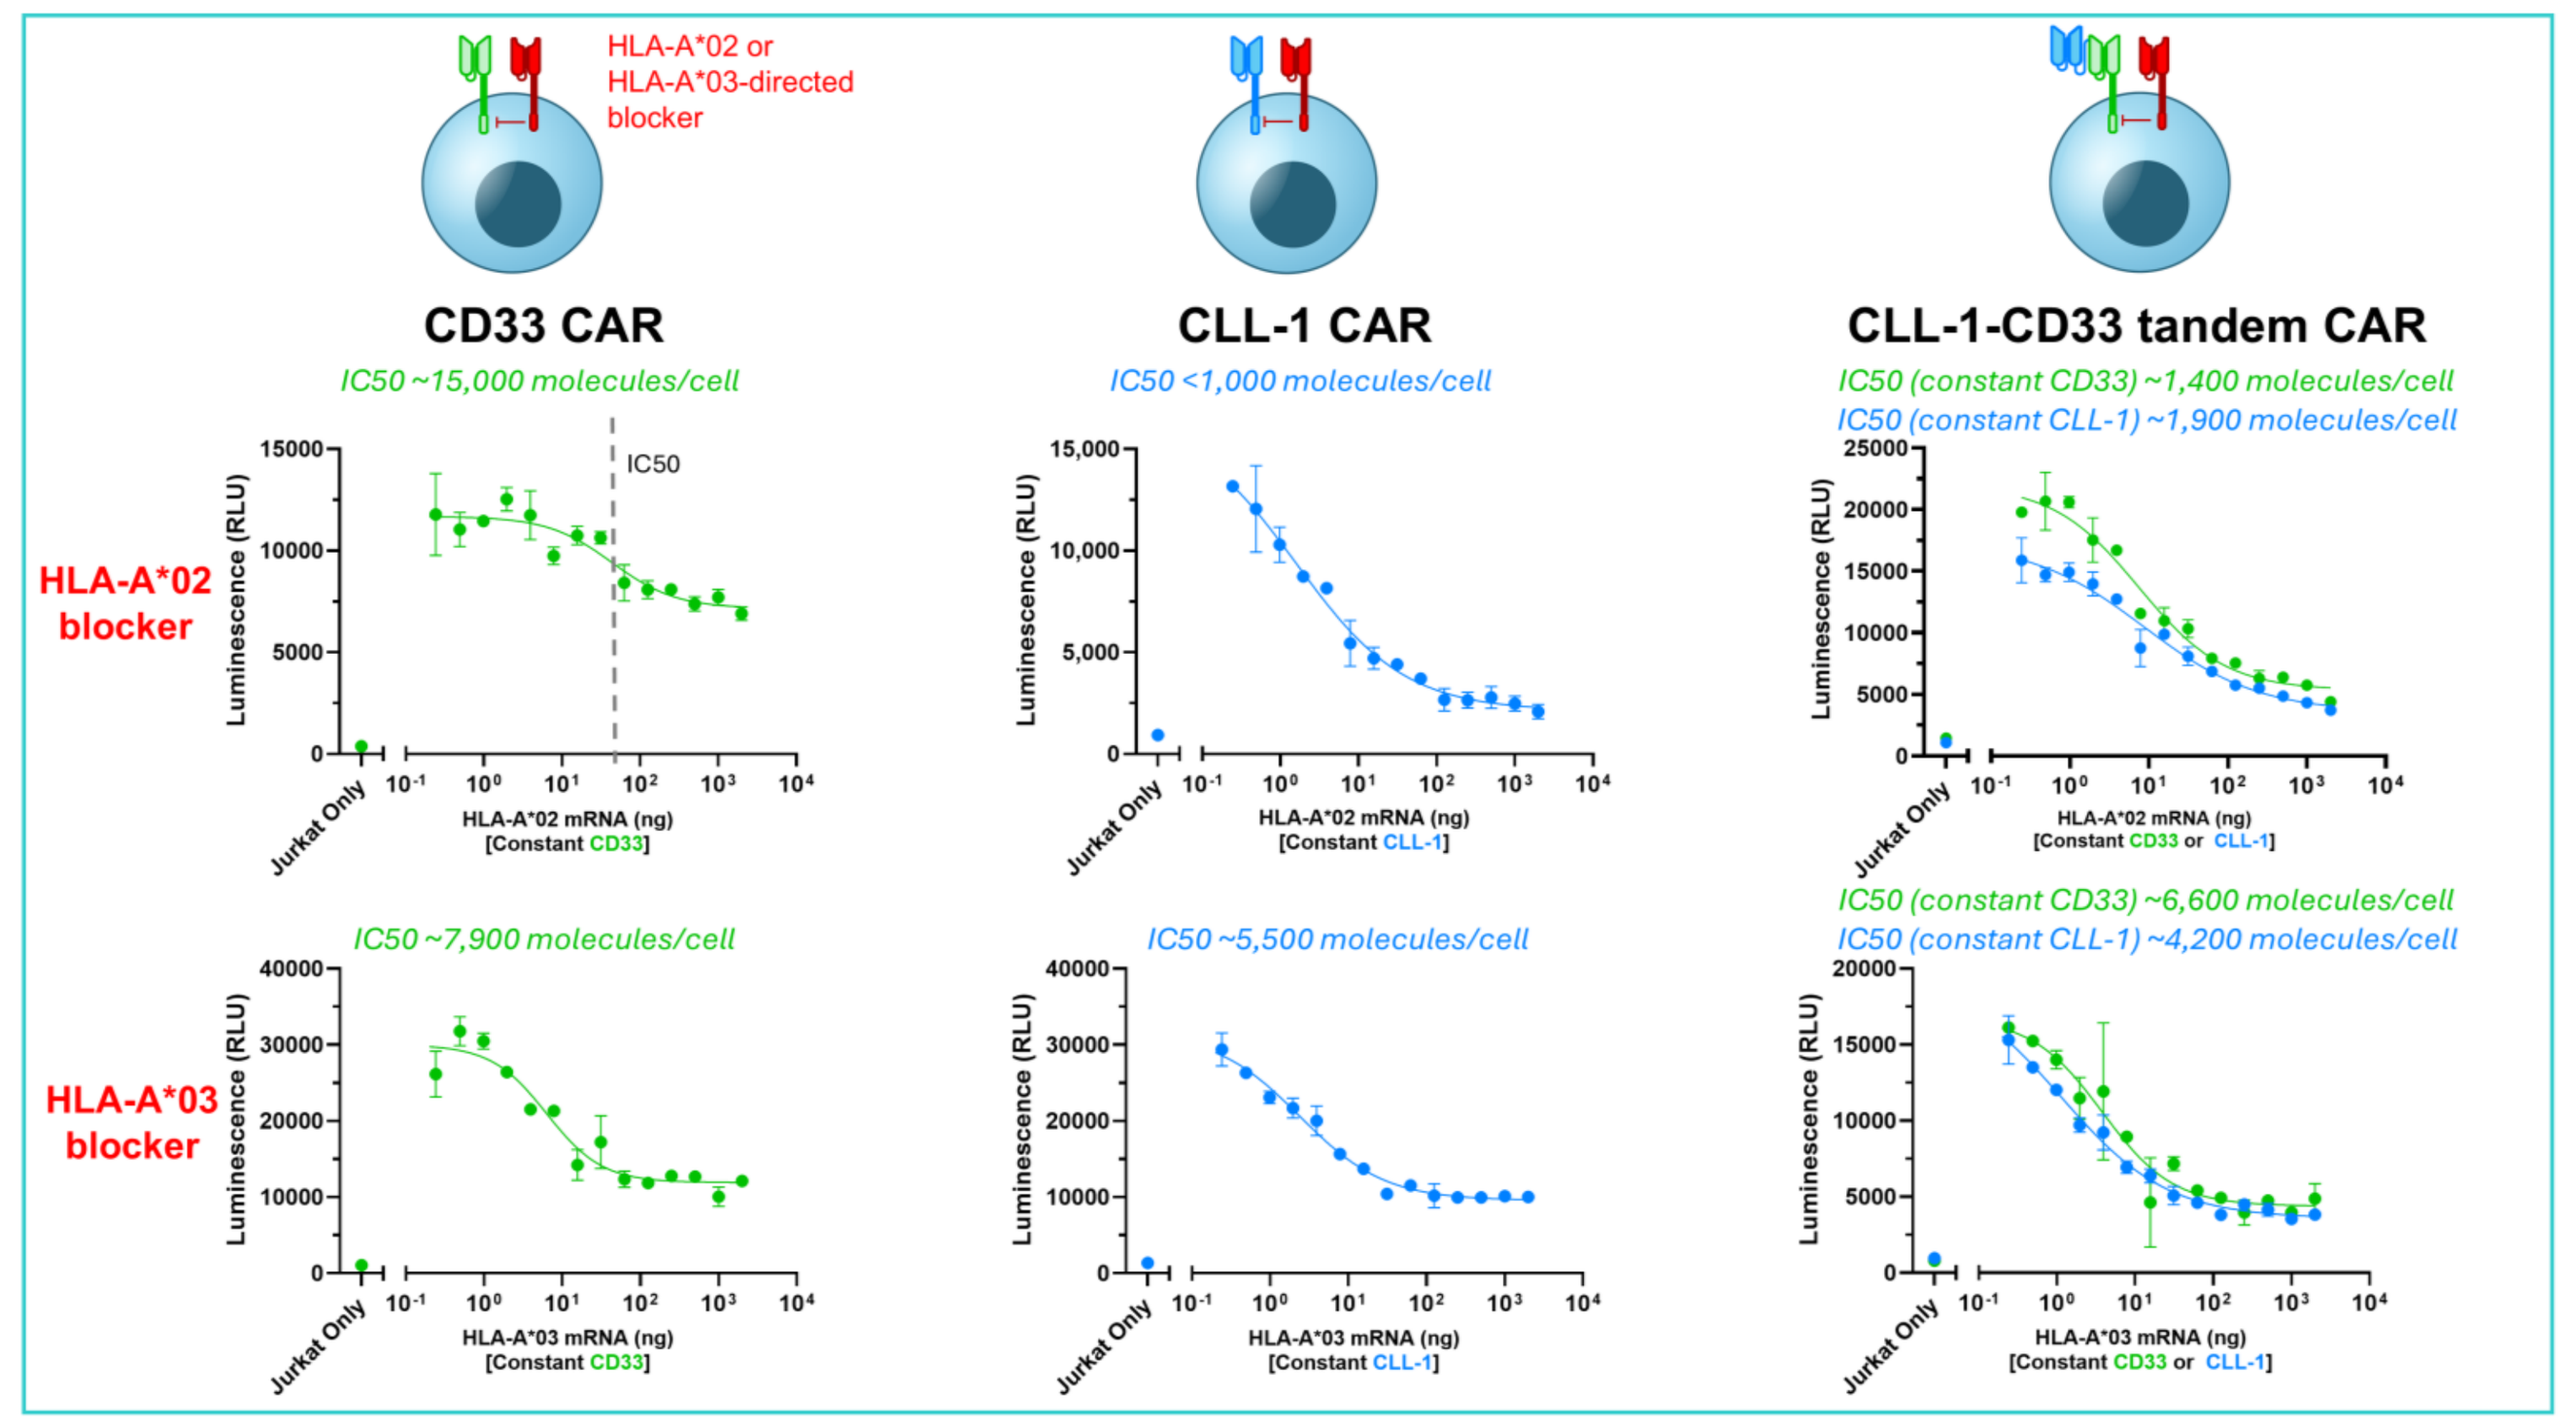
HLA-A*02 or HLA-A*03-directed blocker
CD33 CAR
CLL-1 CAR
CLL-1-CD33 tandem CAR
HLA-A*02 blocker
IC50 ~15,000 molecules/cell
IC50 <1,000 molecules/cell
IC50 (constant CD33) ~1,400 molecules/cell
IC50 (constant CLL-1) ~1,900 molecules/cell
HLA-A*03 blocker
IC50 ~7,900 molecules/cell
IC50 ~5,500 molecules/cell
IC50 (constant CD33) ~6,600 molecules/cell
IC50 (constant CLL-1) ~4,200 molecules/cell
Luminescence (RLU)
HLA-A*02 mRNA (ng)
HLA-A*03 mRNA (ng)

Figure 4: Monospecific CD33 (left) or CLL-1 (middle) CAR as well as a tandem CLL-1-CD33 CAR (right) activation can all be blocked by MHC class I-targeting blockers in Jurkat cells; specifically, an HLA-A*02 blocker (top) and an HLA-A*03 blocker (bottom). Jurkat cells were transiently transfected with CAR constructs and co-cultured with K562 target cells transfected with constant activator antigen mRNA (CD33 or CLL-1) and titrated blocker antigen mRNA (HLA-A*02 or HLA-A*03). NFAT activation was measured via luciferase reporter after 6-hour co-culture of Jurkat and K562 cells.

CLL-1-CD33 | HLA-A*02 TMOD SELECTIVELY KILLS TUMOR CELLS & SPARES PRIMARY HSPCS

The tandem CLL-1-CD33 CAR and HLA-A*02 blocker were expressed in primary T cells via a single lentiviral construct (**Figure 5A**). CLL-1-CD33 Tmod cells demonstrated cytotoxicity against HL-60 target cells expressing CD33 and/or CLL-1, and reduced cytotoxicity against cells expressing HLA-A*02 (**Figure 5B**), including protection of primary HSPCs (**Figure 5C**).

Figure 5: CLL-1-CD33 | HLA-A*02 Tmod demonstrates selective killing of HL-60 target cells, & protection of HLA-A*02+ HL-60 target cells and primary HSPCs

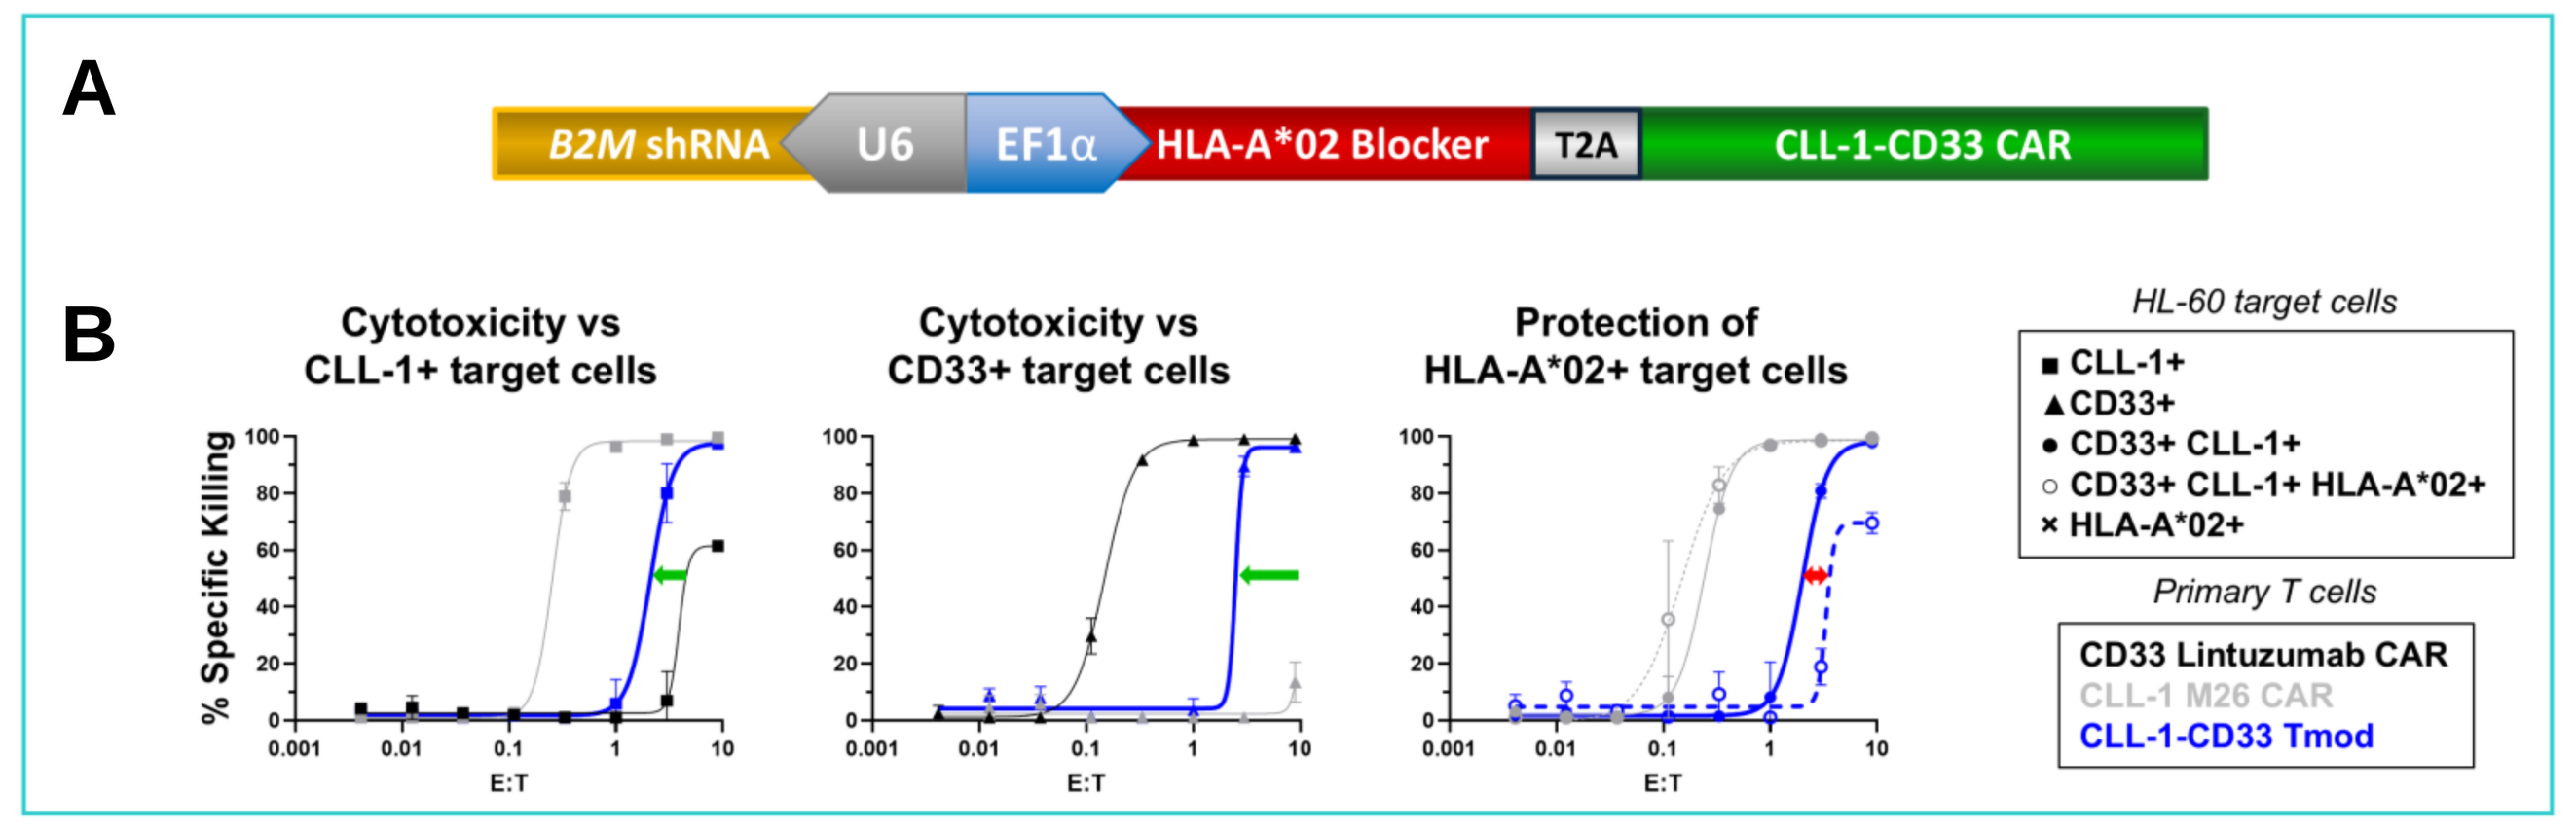
A
B2M shRNA
U6
EF1α
HLA-A*02 Blocker
T2A
CLL-1-CD33 CAR
B
Cytotoxicity vs CLL-1+ target cells
Cytotoxicity vs CD33+ target cells
Protection of HLA-A*02+ target cells
% Specific Killing
E:T
HL-60 target cells
CLL-1+
CD33+
CD33+ CLL-1+
CD33+ CLL-1+ HLA-A*02+
HLA-A*02+
Primary T cells
CD33 Lintuzumab CAR
CLL-1 M26 CAR
CLL-1-CD33 Tmod

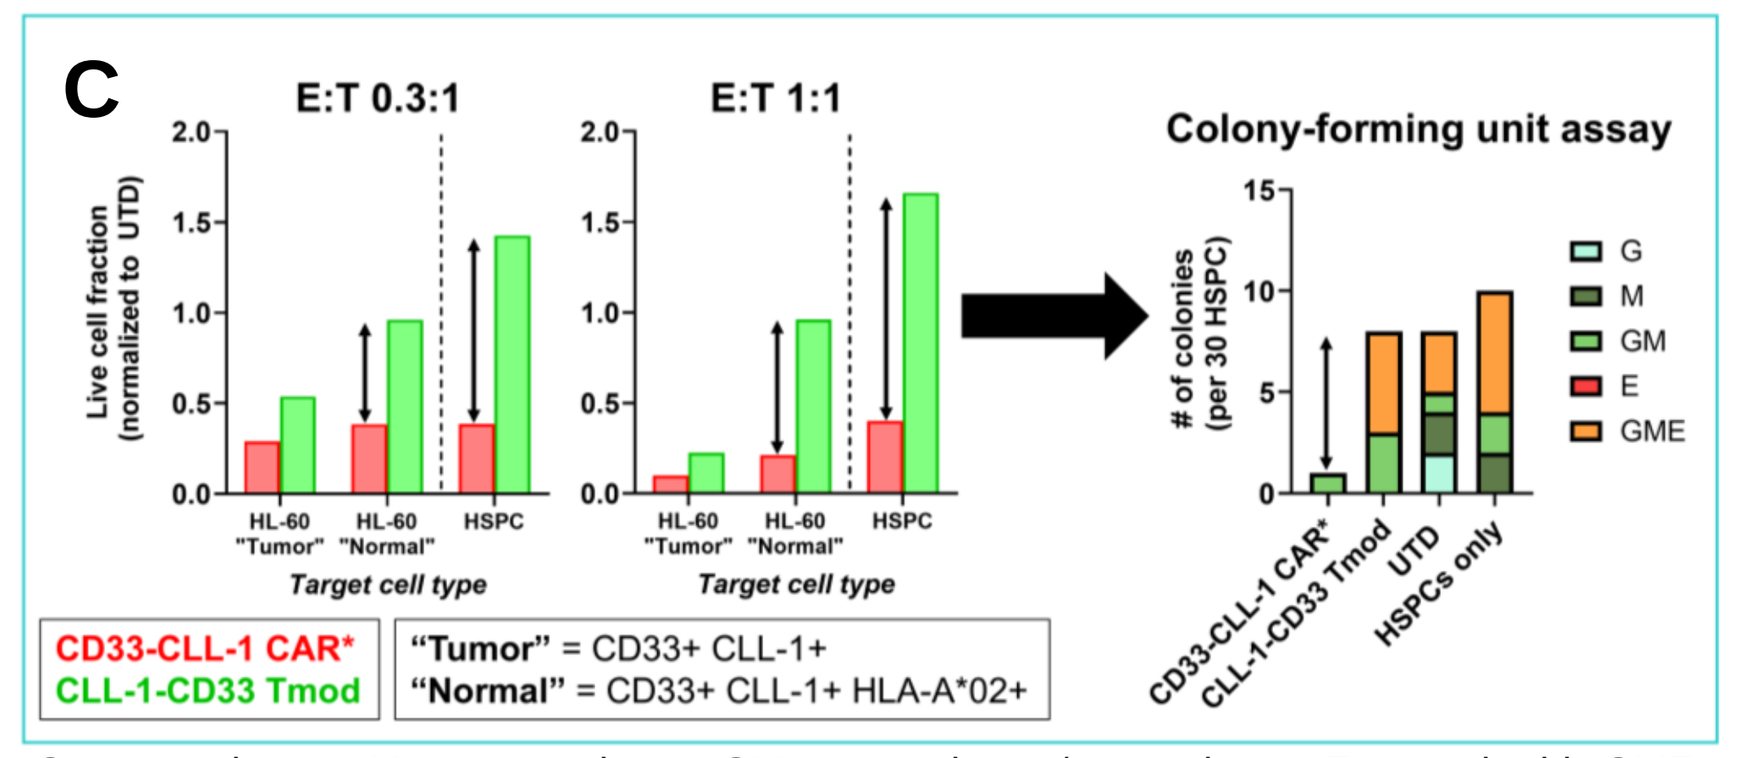
C
E:T 0.3:1
E:T 1:1
Live cell fraction (normalized to UTD)
Target cell type
Colony-forming unit assay
of colonies (per 30 HSPC)
G
M
GM
E
GME
CD33-CLL-1 CAR*
CLL-1-CD33 Tmod
UTD
HSPCs only
"Tumor" = CD33+ CLL-1+
"Normal" = CD33+ CLL-1+ HLA-A*02+

Figure 5: (A) Schematic of a single lentiviral construct encoding (i) an HLA-A*02 blocker, (ii) a tandem CLL-1-CD33 CAR, and (iii) a beta-2 microglobulin (B2M)-directed shRNA. (B) Cytotoxicity of HLA-A*02+ primary T cells transduced with the construct shown in (A) against HL-60 target cells engineered to express CD33, CLL-1, and/or HLA-A*02. (C) CD33-CLL-1 CAR or CLL-1-CD33 Tmod cells (HLA-A*02- donor) were co-cultured with HL-60 target cells or HLA-A*02+ primary HSPCs at the indicated E:T ratios. Following this co-culture, HSPCs were assessed for pluripotency in a colony-forming unit assay.

G = granulocyte, M = macrophage, GM = granulocyte/macrophage, E = erythroid, GME = multi-lineage. *CD33-CLL-1 CAR contains the same scFvs as Tmod but in the reverse order.

CLL-1-CD33 CD28 GEN2 CAR IS MORE POTENT THAN CLL-1-CD33 GEN3 CAR IN VITRO

CD28-based second generation CAR versions (CD28 and CD3ζ ICDs) of the CLL-1-CD33 CAR and Tmod were compared to the original third generation CAR versions (CD28, 4-1BB, and CD3ζ ICDs). The CD28 Gen2 Tmod construct exhibited 3-fold greater potency against CD33 and/or CLL-1+ HL-60 target cells than its Gen3 counterpart (**Figure 6A**), as well as increased antigen-dependent expansion (**Figure 6B**).

Figure 6: CD28 Gen2 CAR is more potent than Gen3 CAR in acute & long-term in vitro assays

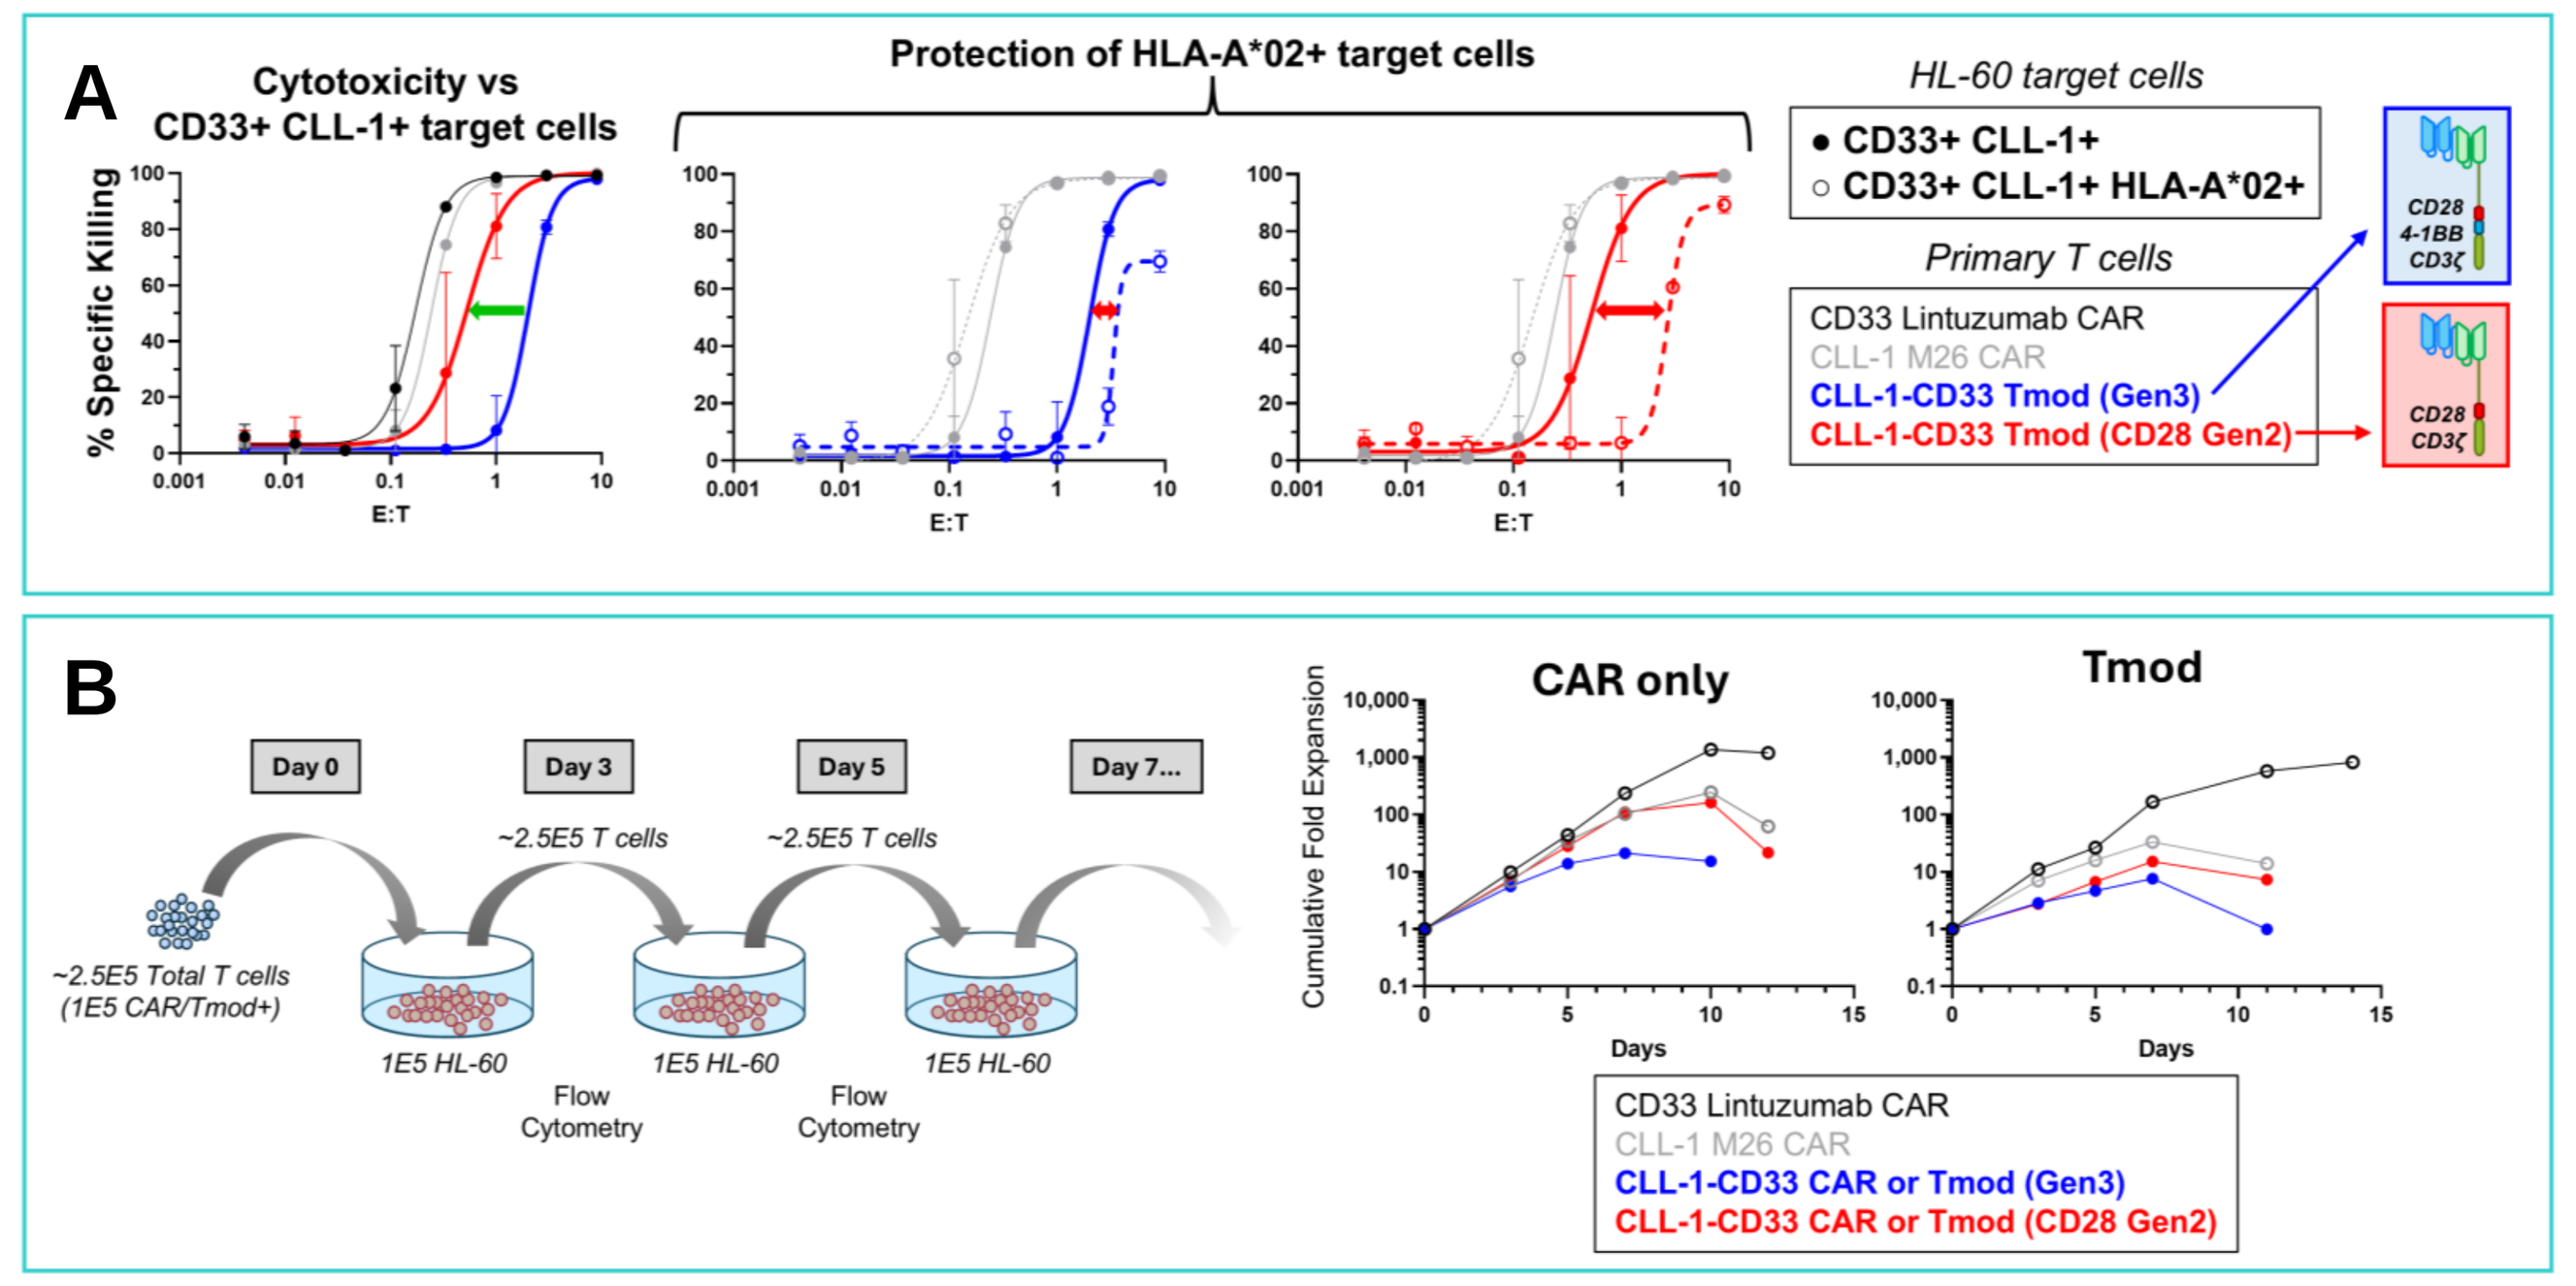
A
Cytotoxicity vs CD33+ CLL-1+ target cells
Protection of HLA-A*02+ target cells
% Specific Killing
E:T
HL-60 target cells
CD33+ CLL-1+
CD33+ CLL-1+ HLA-A*02+
Primary T cells
CD33 Lintuzumab CAR
CLL-1 M26 CAR
CLL-1-CD33 Tmod (Gen3)
CLL-1-CD33 Tmod (CD28 Gen2)
CD28 4-1BB CD3ζ
CD28 CD3ζ
B
Day 0
Day 3
Day 5
Day 7...
~2.5E5 T cells
~2.5E5 Total T cells (1E5 CAR/Tmod+)
1E5 HL-60
Flow Cytometry
CAR only
Tmod
Cumulative Fold Expansion
Days
CD33 Lintuzumab CAR
CLL-1 M26 CAR
CLL-1-CD33 CAR or Tmod (Gen3)
CLL-1-CD33 CAR or Tmod (CD28 Gen2)

Figure 6: (A) A CD28 Gen2 CAR version of CLL-1-CD33 Tmod demonstrated improved potency (~3x) and similar selectivity to a Gen3 CAR version. (B) The same CD28 Gen2 CAR/Tmod has increased antigen-dependent expansion in a repeat antigen challenge assay compared to its Gen3 counterpart. A Lintuzumab-based CD33 CAR and M26-based CLL-1 CAR are shown for comparison.

CLL-1-CD33 CD28 GEN2 TMOD DEMONSTRATES TUMOR CONTROL AND SELECTIVITY IN VIVO

Figure 7: CLL-1-CD33 Tmod (CD28 Gen2) is more potent than CLL-1-CD33 Tmod (Gen3) in an HL-60 xenograft in vivo study

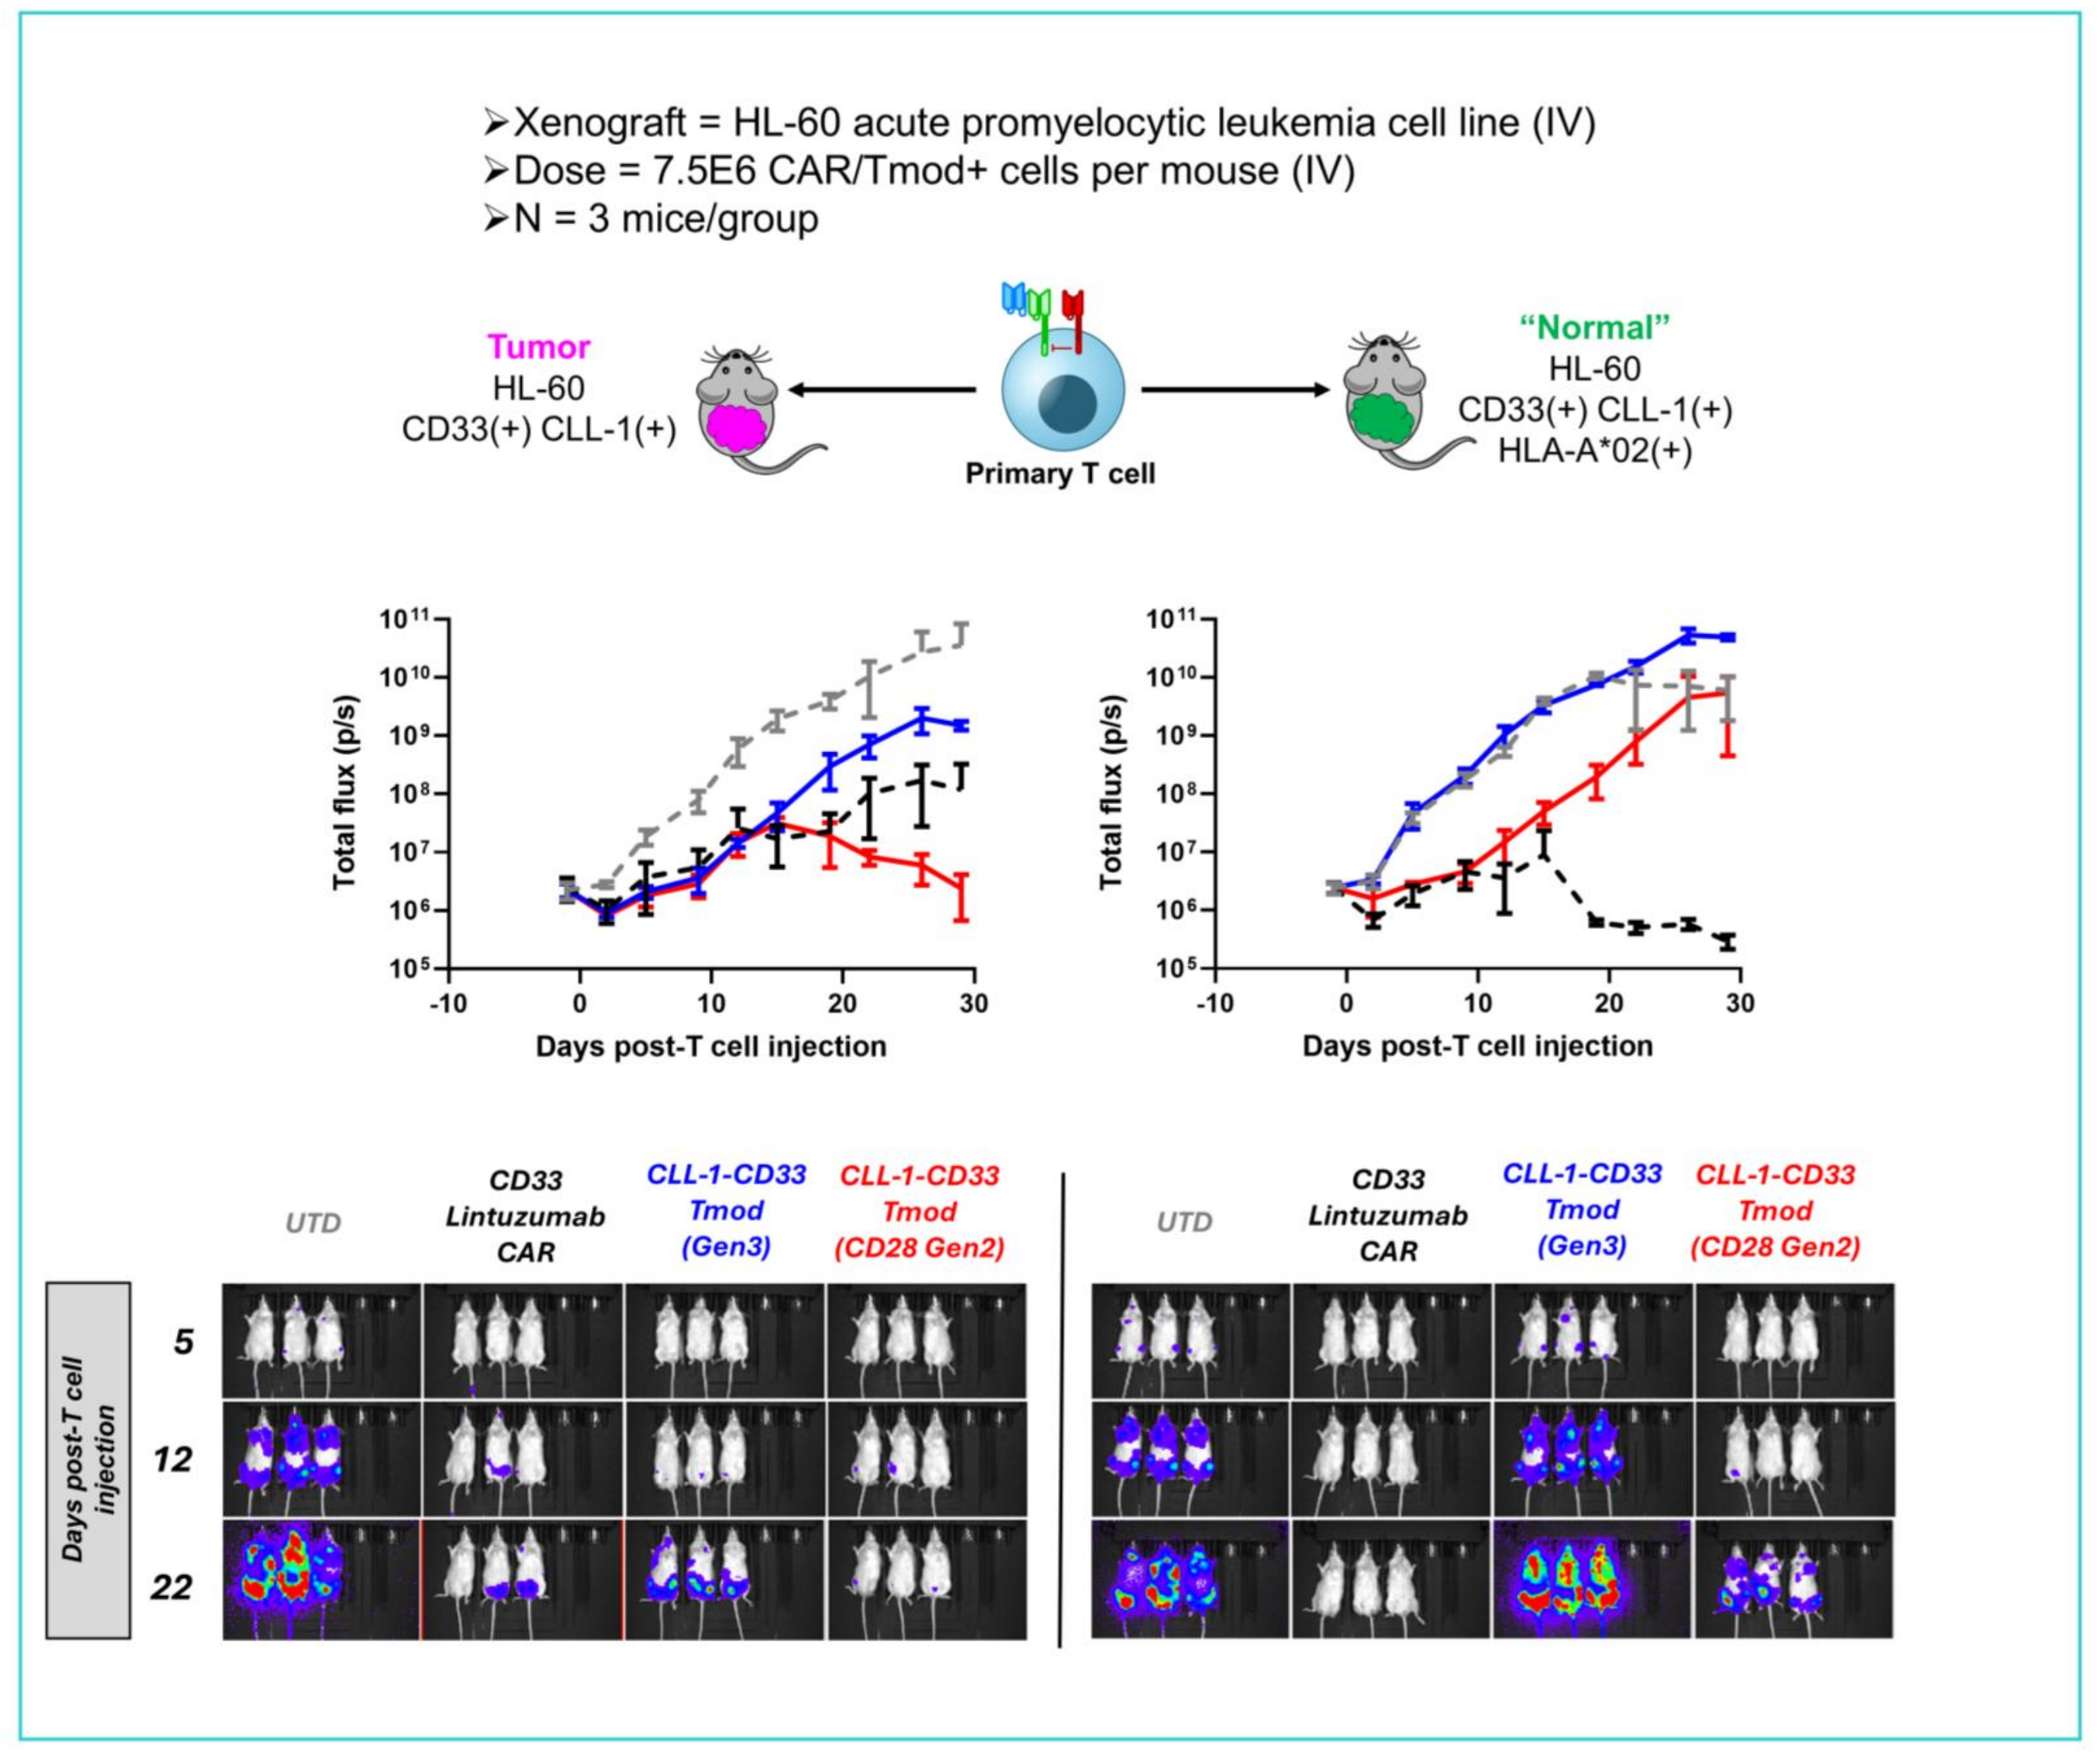
Xenograft = HL-60 acute promyelocytic leukemia cell line (IV)
Dose = 7.5E6 CAR/Tmod+ cells per mouse (IV)
N = 3 mice/group
Tumor
HL-60
CD33(+) CLL-1(+)
Primary T cell
"Normal"
HL-60
CD33(+) CLL-1(+)
HLA-A*02(+)
Total flux (p/s)
Days post-T cell injection
UTD
CD33 Lintuzumab CAR
CLL-1-CD33 Tmod (Gen3)
CLL-1-CD33 Tmod (CD28 Gen2)
Days post-T cell injection
5
12
22

Figure 7: (Top) Schematic of an in vivo xenograft study to evaluate the potency and selectivity of CLL-1-CD33 Tmod. HL-60 target cells were engineered to express firefly luciferase, allowing monitoring of tumor growth via administration of luciferin substrate followed by bioluminescence (BLI) measurements. (Middle) Plots showing BLI measurements of mice with either "tumor" (left) or "normal" (right) xenografts. (Bottom) Images depicting BLI of mice with either "tumor" (left) or "normal" (right) xenografts after treatment with the indicated primary T cells. Images from representative days are shown.

CONCLUSIONS

- CLL-1-CD33 | HLA-A*02 Tmod demonstrates potency against CD33 and/or CLL-1+ cells and protection of HLA-A*02+ cells (including primary HSPCs) in vitro and in vivo
- CD28 Gen2 version of CLL-1-CD33 Tmod is more potent than Gen3 with retained selectivity

References

1. Cancer Genome Atlas Research Network. N Engl J Med. 2013 May 30;368(22):2059-74.
2. Therapeutically Applicable Research to Generate Effective Treatments (TARGET) initiative, phs000218, managed by the NCI. The data used for this analysis are available [https://www.ncbi.nlm.nih.gov/projects/gap/cgi-bin/study.cgi?study_id=phs000465.v21.p8]. Information about TARGET can be found at <http://ocg.cancer.gov/programs/target>.
3. Chen L, et al. Science. 2014 Sep 26;345(6204):1251033.
4. Fares I, et al. Blood, The Journal of the American Society of Hematology. 2017 Jun 22;129(25):3344-51.
5. Tomellin i E, et al. Cell Rep. 2019 Jul 23;28(4):1063-1073.e5.
6. Solman M, et al. Elife. 2022 May 10;11:e73040.
7. Monaco G, et al. Cell Rep. 2019;26(6):1627-1640.e7.
8. Ghandi M, et al. Nature. 2019 May 08. 569, 503–508.
9. Hamburger AE, et al. Mol Immunol. 2020;128:298 310.

Presented at the American Association for Cancer Research Annual Meeting, April 25-30, 2025

dmanry@a2biotherapeutics.com