Defining the Biophysical Design Parameters for Logic-Gated CAR-T Systems

Alexander C. Partin[1], Richele Bruno[1], Sanam Shafaattalab[1], Erica Vander Mause[1], Aaron Winters[1], Mark Daris[1], Casey Gahrs[1], Claudia A. Jette[1], Breanna DiAndreth[1], Mark L. Sandberg[1], Agnes E. Hamburger[1], Alexander Kamb[1], Timothy P. Riley[1]

*[1]Discovery Research, A2 Biotherapeutics, Inc., 30301 Agoura Road, Agoura Hills, CA 91301

Summary and Key Points

- Logic-gated, two-receptor CAR-T systems, such as Tmod™, are a promising strategy to enforce tumor selectivity and overcome on-target/off-tumor toxicities, but pose new considerations for receptor design
- To inform design goals for expansion of Tmod targets, we characterized how antigen geometry and epitope affinity influence blocking efficiency
- Tmod blockers show optimal blocking against small, membrane-proximal antigens, but a broader range of antigens can be targeted effectively through the use of engineered rigid hinges
- Systematic rigid hinge characterization was used to identify the the geometric parameters defining optimal activator/blocker pairs
- In vitro characterization of affinity-attenuated FLAG sequence variants indicate that affinity plays a role in both activation and blocking in Tmod systems

Introduction

Engineered immune cells, such as CAR-T cells, show promise for immuno-oncology but must overcome the obstacle of on-target/off-tumor toxicity. Novel strategies, like synthetic Notch receptors and the Tmod platform, offer promising avenues by recognizing combinatorial antigen profiles. The modular Tmod system integrates an activating CAR with an inhibitory receptor, providing a safety switch to spare normal tissues expressing inhibitory antigens.

The first generation of Tmod blockers targets the HLA-A*02 antigen, which is lost in certain tumors via loss-of-heterozygosity. Expanding Tmod to a broader patient population will require the design of tailored blocker modules for diverse antigens. In this work we aimed to characterize the determinants for optimal blocking, to better define the design guidelines for the next generation of Tmod blocker modules. We reveal the impact of antigen geometry and affinity on blocking potency, and develop an engineering approach to improving blocking via modulating geometric parameters of activator and blocker pairs.

Figure 1: Tmod logic-gated CAR-T cells for targeting distinct antigen profiles

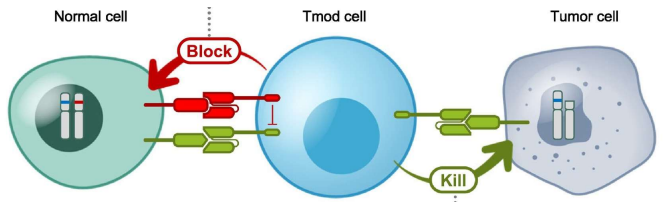
Normal cell
Tmod cell
Tumor cell
Block
Kill

*Tmod cells are designed to selectively kill tumor cells based on a distinct antigen profile. Normal cells expressing a blocker antigen (HLA-A*02 in the first generation of Tmod) preclude activation via an inhibitory receptor, whereas tumor cells lacking the blocker antigen trigger activation and killing by the Tmod cell .* From DiAndreth et al.[2]

Expanding Tmod beyond HLA-A*02

- Blockers targeting HLA-A*02, MSLN, and ICAM-1 antigens were developed to block the activation signal from a CD19 CAR
- HLA-A*02 induced higher inhibition levels compared to those targeting MSLN and ICAM-1

Figure 2: Blocking efficiencies vary widely for different blocker/target pairs

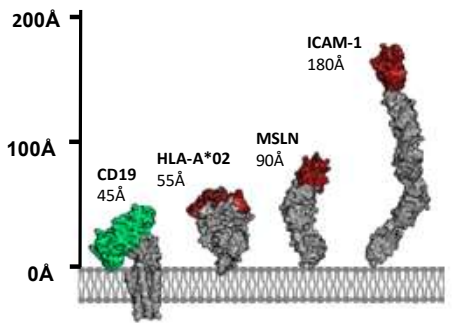
200Å
100Å
0Å
CD19
45Å
HLA-A*02
55Å
MSLN
90Å
ICAM-1
180Å

*Structural comparison of CD19 A-Antigen (epitope in green) and three B-Antigens (HLA-A*02, MSLN, and ICAM-1; epitopes in red).*

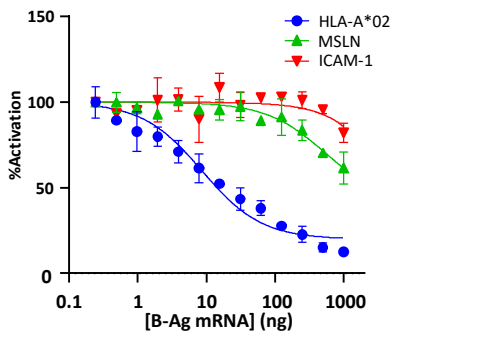
HLA-A*02
MSLN
ICAM-1
%Activation
150
100
50
0
0.1
1
10
100
1000
[B-Ag mRNA] (ng)

Jurkat T cells expressing the CD19 activator and the blocker indicated in the legend, co-cultured with K562(CD19+) target cells transfected with titration of B-antigen mRNA.

HLA-A*02 (K_D: 190nM) **MSLN** (K_D: 30nM) **ICAM-1** (K_D: 39nM)

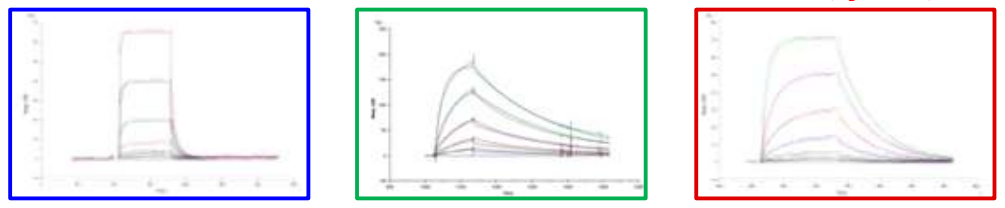

Surface plasmon resonance sensorgram measurements of monovalent affinities for blocker antigens shown in Figure 2. No correlation was observed for affinity and blocking efficiency.

Blocker antigen size influences inhibitory function

- Blocking efficiency of a FLAG-specific blocker was measured using an NFAT reporter system
- Anti-FLAG blocker function is inversely related to antigen size

Figure 3: FLAG-based blocking assays reveal the impact of blocker antigen size

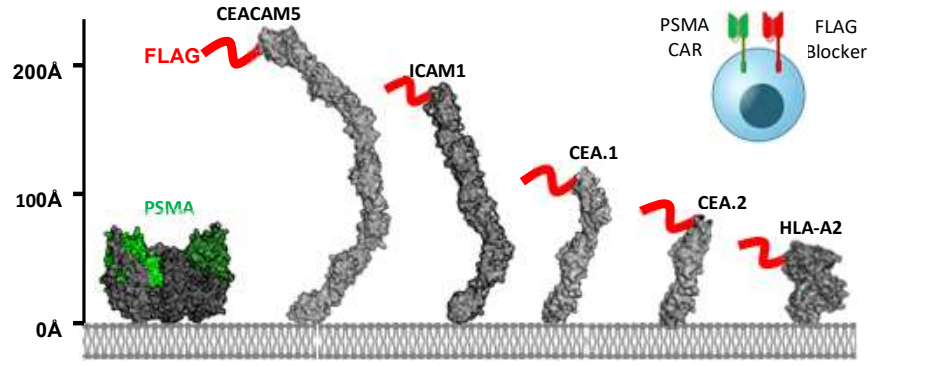
200Å
100Å
0Å
FLAG
PSMA
CEACAM5
ICAM1
CEA.1
CEA.2
HLA-A2
PSMA CAR
FLAG Blocker

FLAG-tagged antigens of different sizes were evaluated for their blocking efficiencies when paired with PSMA activator CAR in Jurkat-based NFAT reporter assay.

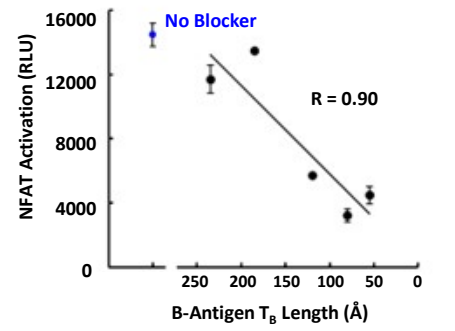
NFAT Activation (RLU)
16000
12000
8000
4000
0
No Blocker
R = 0.90
250
200
150
100
50
0
B-Antigen T_B Length (Å)

Relationship between blocker antigen axial length and blocking efficiency in a Jurkat-based NFAT reporter assay.

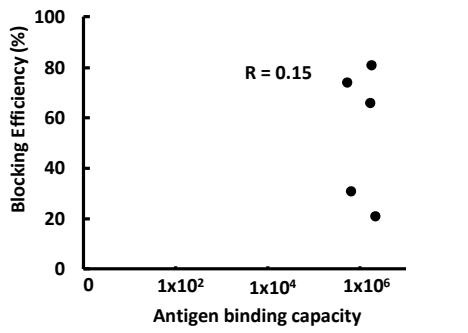
Blocking Efficiency (%)
100
80
60
40
20
0
R = 0.15
0
1x10²
1x10⁴
1x10⁶
Antigen binding capacity

Relationship between blocker antigen axial length and blocking efficiency in a Jurkat-based NFAT reporter assay.

Modular hinges for controlling activator and blocker geometries

Figure 4: Design of semi-rigid modular EGF hinges to manipulate effector-target interface

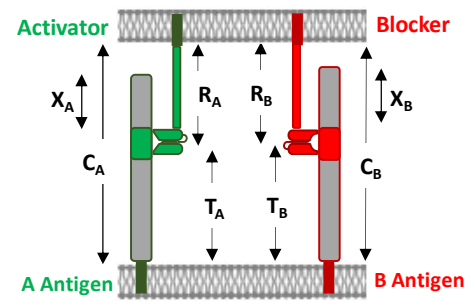
Activator
Blocker
A Antigen
B Antigen

Activator and blocker complexes were characterized by their geometric parameters

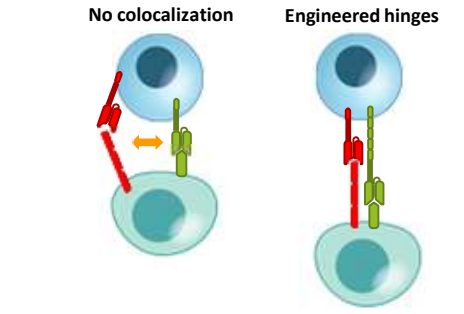
No colocalization
Engineered hinges

Manipulating activator and blocker modules could enforce compatible activator/blocker geometries

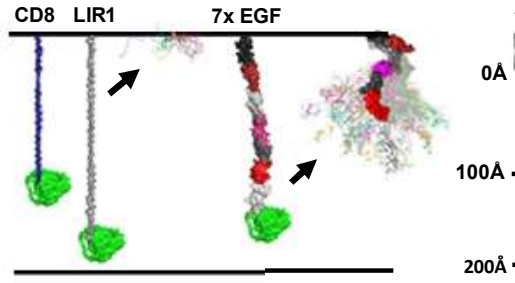
CD8
LIR1
7x EGF

Relative flexibilities of Tmod platform hinges for activator and blocker (CD8 and LIR1, respectively) versus semi-rigid EGF hinge. 100 Monte Carlo simulations were overlayed to assess flexibility.

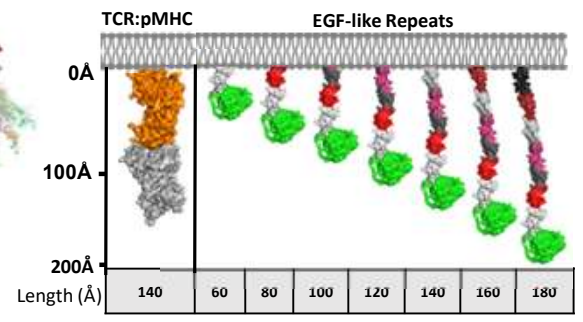
TCR:pMHC
EGF-like Repeats
0Å
100Å
200Å
Length (Å)
140
60
80
100
120
140
160
180

Comparison of dimensions for TCR:pMHC complex and a series of semi-rigid EGF rigid hinge designs. Each EGF module is estimated to contribute an additional 20-30 Å to axial height of hinge.

Tuning Tmod with elongated hinges

- Modulating activator and blocker hinges can reveal optimal geometry
- Longer activators paired with shorter blockers demonstrated maximal blocking efficiencies in both Jurkat and primary T cell assays

Figure 5. Functional evaluation of engineered rigid hinge combinations

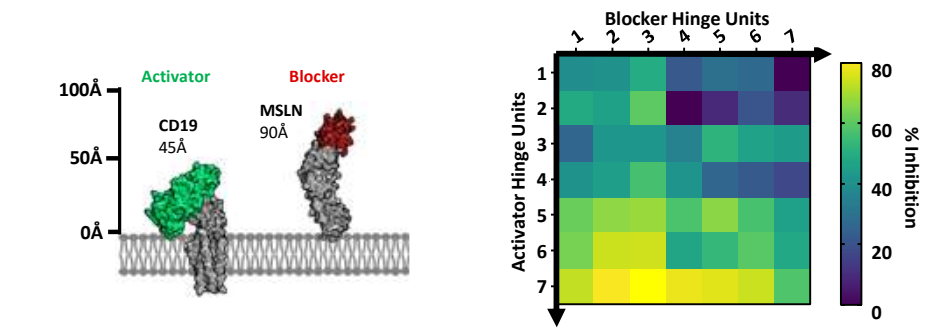
100Å
50Å
0Å
Activator
Blocker
CD19
45Å
MSLN
90Å
Blocker Hinge Units
Activator Hinge Units
% Inhibition

Jurkat NFAT cells expressing each hinge combination of CD19 activator plus MSLN blocker were co-cultured with K562(MSLN-) or K562(MSLN+) cells. MSLN-dependent NFAT activation was converted into blocking efficiency (%) and presented as a heatmap.

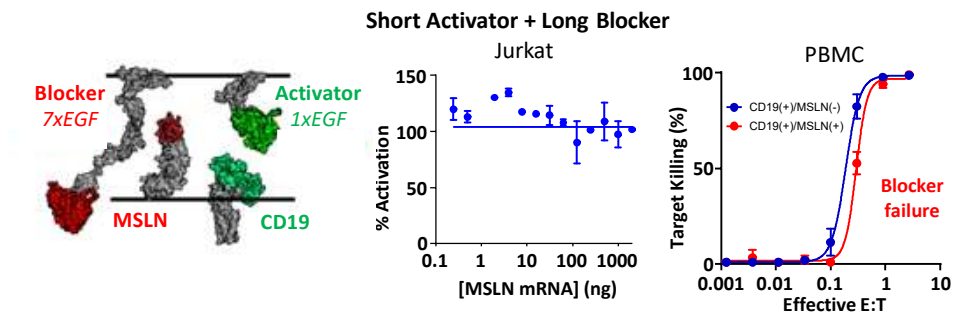
Short Activator + Long Blocker
Blocker
7xEGF
Activator
1xEGF
MSLN
CD19
Jurkat
% Activation
[MSLN mRNA] (ng)
PBMC
Target Killing (%)
CD19(+)/MSLN(-)
CD19(+)/MSLN(+)
Blocker failure
Effective E:T

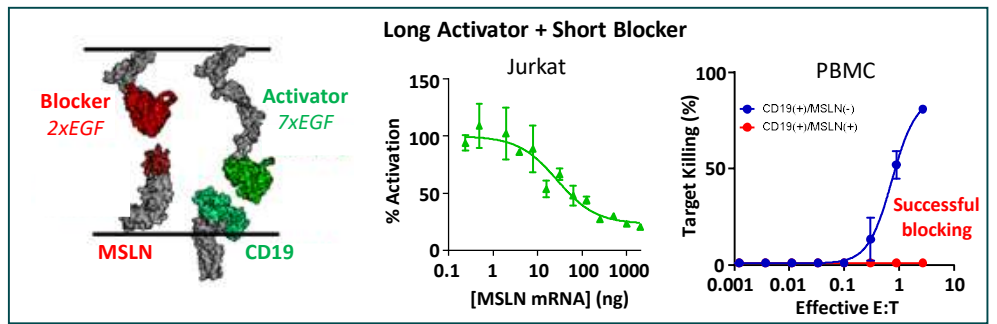
Long Activator + Short Blocker
Blocker
2xEGF
Activator
7xEGF
MSLN
CD19
Jurkat
% Activation
[MSLN mRNA] (ng)
PBMC
Target Killing (%)
CD19(+)/MSLN(-)
CD19(+)/MSLN(+)
Successful blocking
Effective E:T

Left, Schematics illustrating hypothesized activator/blocker dimensions and compatibilities. Center, Jurkat-based NFAT blocking assays using hinge configurations indicated in schematic. Right, cytotoxicity assays of PBMCs transfected with hinge configurations at a series of Effector:Target ratios.

Engineered hinges reveal geometric parameters for optimal blocking

- Additional rigid hinge datasets were generated using the antigens shown in Figure 3.
- The differences between blocker complex size and activator complex size ($\Delta C_{B\text{-}A}$) were estimated using structural modeling, and plotted against blocking efficiency.

Figure 6. Expanding rigid hinge analysis to other targets reveals a geometric trend

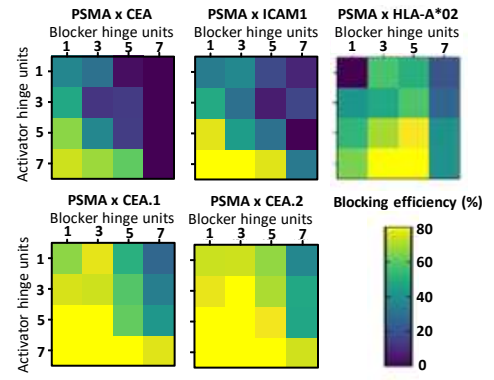
PSMA x CEA
PSMA x ICAM1
PSMA x HLA-A*02
PSMA x CEA.1
PSMA x CEA.2
Blocker hinge units
Activator hinge units
Blocking efficiency (%)

Rigid hinge matrix heatmaps showing blocking efficiencies for indicated Activator/Blocker hinge combinations, for FLAG-tagged blocker antigens depicted in Figure 3.

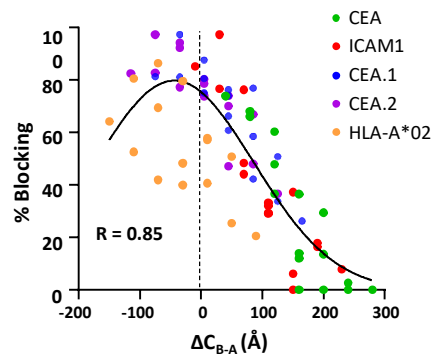
CEA
ICAM1
CEA.1
CEA.2
HLA-A*02
% Blocking
R = 0.85
$\Delta C_{B\text{-}A}$ (Å)

Correlation between maximal inhibition (Imax, %) and the estimated difference in complex length between blocker and activator ($\Delta C_{B\text{-}A}$) using modular EGF hinges.

Correlating binding strength with activation and blocking

- A series of FLAG tag mutants interact with anti-FLAG activators and blockers with differing affinities
- Affinity-dependent activation and blocking efficiencies were observed for each FLAG target variant

Figure 7. Design and characterization of FLAG tag variants with attenuated affinities

FLAG Variant									ELISA Binding[5]
DFKDDDDK	D	F	K	D	D	D	D	K	0.2
EYKDDDDK	E	Y	K	D	D	D	D	K	0.5
DYKDDADK	D	Y	K	D	D	A	D	K	0.9
DYKVVDDR	D	Y	K	V	V	D	D	R	1.4
DYKDDDAK	D	Y	K	D	D	D	A	K	1.8
DYKEDDDK	D	Y	K	E	D	D	D	K	2.2
DYKDDEDK	D	Y	K	D	D	E	D	K	2.4
DYKDDDEK	D	Y	K	D	D	D	E	K	2.7
DYKDEDDK	D	Y	K	D	E	D	D	K	2.8
"WT" DYKDDDDK	D	Y	K	D	D	D	D	K	3
DYKDDDDR	D	Y	K	D	D	D	D	R	3.2

Table showing FLAG variants selected for conversion to blocker antigen epitopes. ELISA binding to anti-FLAG (M2) antibody for each variant are indicated. Conventional FLAG sequence ("WT") is indicated. From Slootstra et al.

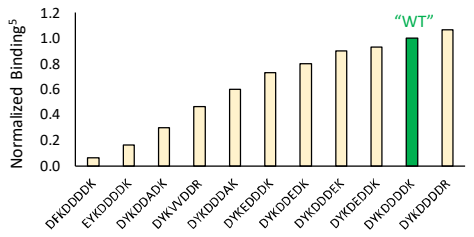
Normalized Binding[5]
"WT"
DFKDDDDK
EYKDDDDK
DYKDDADK
DYKVVDDR
DYKDDDAK
DYKEDDDK
DYKDDEDK
DYKDDDEK
DYKDEDDK
DYKDDDDK
DYKDDDDR

Normalized binding to anti-FLAG (M2) for each FLAG tag variant. A total of 11 variants were selected based on achieving a broad range of affinities for Tmod studies. From Slootstra et al.

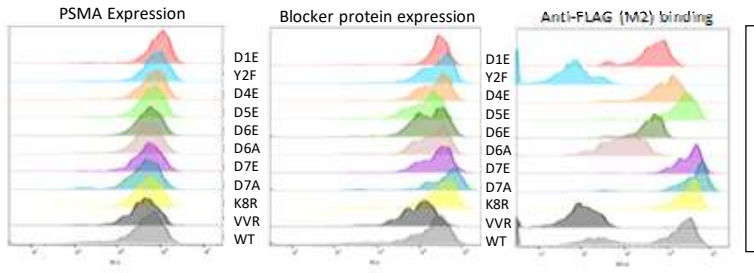
PSMA Expression
Blocker protein expression
Anti-FLAG (M2) binding
D1E
Y2F
D4E
D5E
D6E
D6A
D7E
D7A
K8R
VVR
WT

Flow cytometry data showing similar expression levels for PSMA (left) and a FLAG tagged dummy protein (center). As predicted[5], FLAG mutants show a broad range of binding behaviors when stained with anti-FLAG (M2) antibody (right).

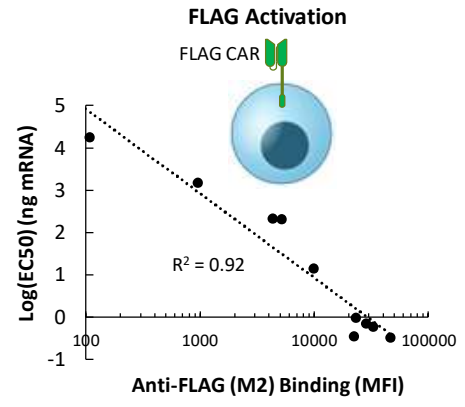
FLAG Activation
FLAG CAR
Log(EC50) (ng mRNA)
R^2 = 0.92
Anti-FLAG (M2) Binding (MFI)

Correlation between anti-FLAG(M2) binding and activation sensitivity (EC50) in Jurkat-based NFAT reporter assay.

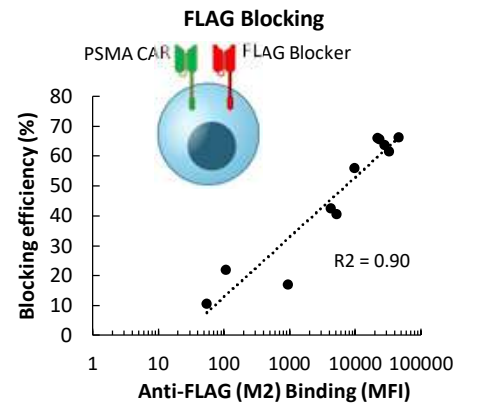
FLAG Blocking
PSMA CAR
FLAG Blocker
Blocking efficiency (%)
R2 = 0.90
Anti-FLAG (M2) Binding (MFI)

Correlation between anti-FLAG(M2) binding and blocking efficiency (%) in Jurkat-based NFAT reporter assay.

Discussion

The challenge of achieving tumor selectivity in cancer therapies underscores the need for novel targeting mechanisms. The Tmod approach, leveraging a NOT gated CAR to target tumor-specific antigen profiles, presents a promising avenue. However, the introduction of a second receptor as a blocker module poses new obstacles, including complexities in design and co-expression, as well as challenges in targeting pairs of diverse antigens. We explored how tuning receptor dimensions influences synapse formation and found that rational receptor design can compensate for suboptimal conditions. Tuning affinity may also provide a lever to improve integration of diverse antigens into logic-gated CARs. While our study provides a framework for optimizing logic-gated T cell therapeutics, further work is needed to validate our in vitro observations in vivo and in the clinic.

Our findings suggest that the Tmod platform is highly versatile and can demonstrate tumor selectivity in a wide range of contexts beyond HLA. This work describes a strategy for optimizing receptor co-localization, which is broadly applicable to logic-gated CARs or other dual-receptor systems. While these findings represent a step forward in addressing on-target/off-tumor toxicity, a more comprehensive understanding of synthetic logic-gated systems is needed for cancer cell therapy to overcome this and other longstanding obstacles.

References

1. Hamburger *et al.,* Engineered T cells directed at tumors with defined allelic loss. *Mol Immunol* 2020
2. DiAndreth *et al.,* The Tmod cellular logic gate as a solution for tumor-selective immunotherapy. *Clin. Immunol 2022*
3. Tokatlian *et al.,* Mesothelin-specific CAR-T cells therapy that incorporates an HLA-gated safety mechanism selectively kills tumor cells. *J Immunother Cancer* 2022
4. Peng *et al.,* Mass Spectrometry-based de novo sequencing of monoclonal antibodies using multiple proteases and a dual fragmentation scheme. *J. Proteome Res. 2021*
5. Slootstra *et al.,* Identification of new tag sequences with differential and selective recognition properties for the anti-FLAG monoclonal antibodies M1, M2 and M5. *Mol Divers. 1997*
6. Partin *et al.*, Geometric parameters that affect the behavior of logic-gated CAR T cells. Front. Immunol. 2024