Abstract Number 77

An NGS assay to identify HLA-A loss of heterozygosity for future CEA and MSLN logic-gated CAR-T solid tumor protocols designed for reduced on-target, off-tumor toxicity

Scott Kopetz[1], M. Pia Morelli[1], Julian R. Molina[2], Diane M. Simeone[3], J. Randolph Hecht[4], Kedar Kirtane[5], Mitesh J. Borad[6], Theodore H. Welling[3], Edward Garon[4], Armen Mardiros[7], Xueyin Wang[7], Eric W. Ng[7], Tyler Danek[8], Shannon Gallagher[8], Ariane Lozac'hmeur[8], Karl Beutner[8], John S. Welch[7], David Maloney[9], William Y. Go[7], Sandip P. Patel[10]

[1]Department of Gastrointestinal Medical Oncology, Division of Cancer Medicine, The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [2]Division of Medical Oncology, Mayo Clinic, Rochester, MN, USA; [3]Department of Surgery, New York University Langone Health, New York, NY, USA; [4]David Geffen School of Medicine at University of California at Los Angeles, Los Angeles, CA, USA; [5]H. Lee Moffitt Cancer Center, Tampa, FL, USA; [6]Mayo Clinic Cancer Center, Phoenix, AZ, USA; [7]A2 Biotherapeutics, Inc., Agoura Hills, CA, USA; [8]Tempus, Chicago, IL, USA; [9]Clinical Research Division, Fred Hutchinson Cancer Research Center, Seattle, WA, USA; [10]Department of Medical Oncology, University of California San Diego, San Diego, CA, USA

BACKGROUND

- Survival in relapsed or metastatic solid tumors remains poor. Current 5-year survivals are less than 30% across multiple tumor types, and less than 10% in many cancers (**Table 1**) [1]
- Chimeric antigen receptor (CAR) T-cell therapy has demonstrated clinical outcomes in hematologic malignancies [2,3]. However, translating engineered T-cell therapies to solid tumors proves difficult due to a lack of tumor-specific targets that distinguish cancer cells from normal cells. In previous studies, the use of a carcinoembryonic antigen (CEA) T-cell receptors and mesothelin (MSLN) CARs both resulted in dose-limiting on-target, off-tumor toxicities [4,5]
- Tmod™ CAR T-cell therapy addresses these challenges by leveraging dual receptors to create a robust AND-NOT signal integrator capable of killing tumor cells while leaving healthy cells intact [6]. Tmod platform technology is a versatile system that may be applied to T cells and natural killer cells in autologous and allogeneic settings
- Human leukocyte antigen loss of heterozygosity (HLA LOH) offers a definitive tumor versus normal discriminator target for CAR T-cell therapy [6,7]. The 2 receptors of the Tmod CAR T-cell platform comprise an activator that recognizes an antigen present on the surface of normal and tumor cells and a blocker that recognizes a second surface antigen from an HLA-A allele lost only in tumor cells (**Figure 1**)
 - The first blocker targets HLA-A*02 because this is the most prevalent allele in the US population
- HLA-A LOH has been observed in ~13% of all solid tumors and up to 33% of primary pancreatic cancers, tumor types with significant unmet medical need [8]. New technologies have shown higher HLA-A LOH rates; however, it is unclear whether patients with HLA-A LOH in their primary tumor tissues are at higher risk for recurrence
- Prevalence of HLA-A LOH across gastrointestinal (GI) tumors is unknown in the real-world setting
- The Tempus xT next-generation sequencing (NGS) database of patients with multiple GI tumors can help determine HLA-A LOH advanced disease in the real world
- With the Tempus xT standard-of-care NGS assay, patients with GI cancer can be readily identified for HLA-A LOH and future treatment with Tmod CAR T therapy

Table 1. Low bar for efficacy

High unmet medical need	
Metastatic cancer	**5-year survival[a]**
Colorectal	15%
Gastroesophageal	5%-6%
Pancreatic	3%
NSCLC	8%
Mesothelioma	8%
Head and neck	~23%
Prostate	31%
Ovarian	30%
Breast	29%

[a] Adapted from The American Cancer Society [1]
NSCLC, non-small cell lung cancer.

Figure 1. Logic-gated CAR to reduce toxicity: activator and HLA-A*02 blocker strategy

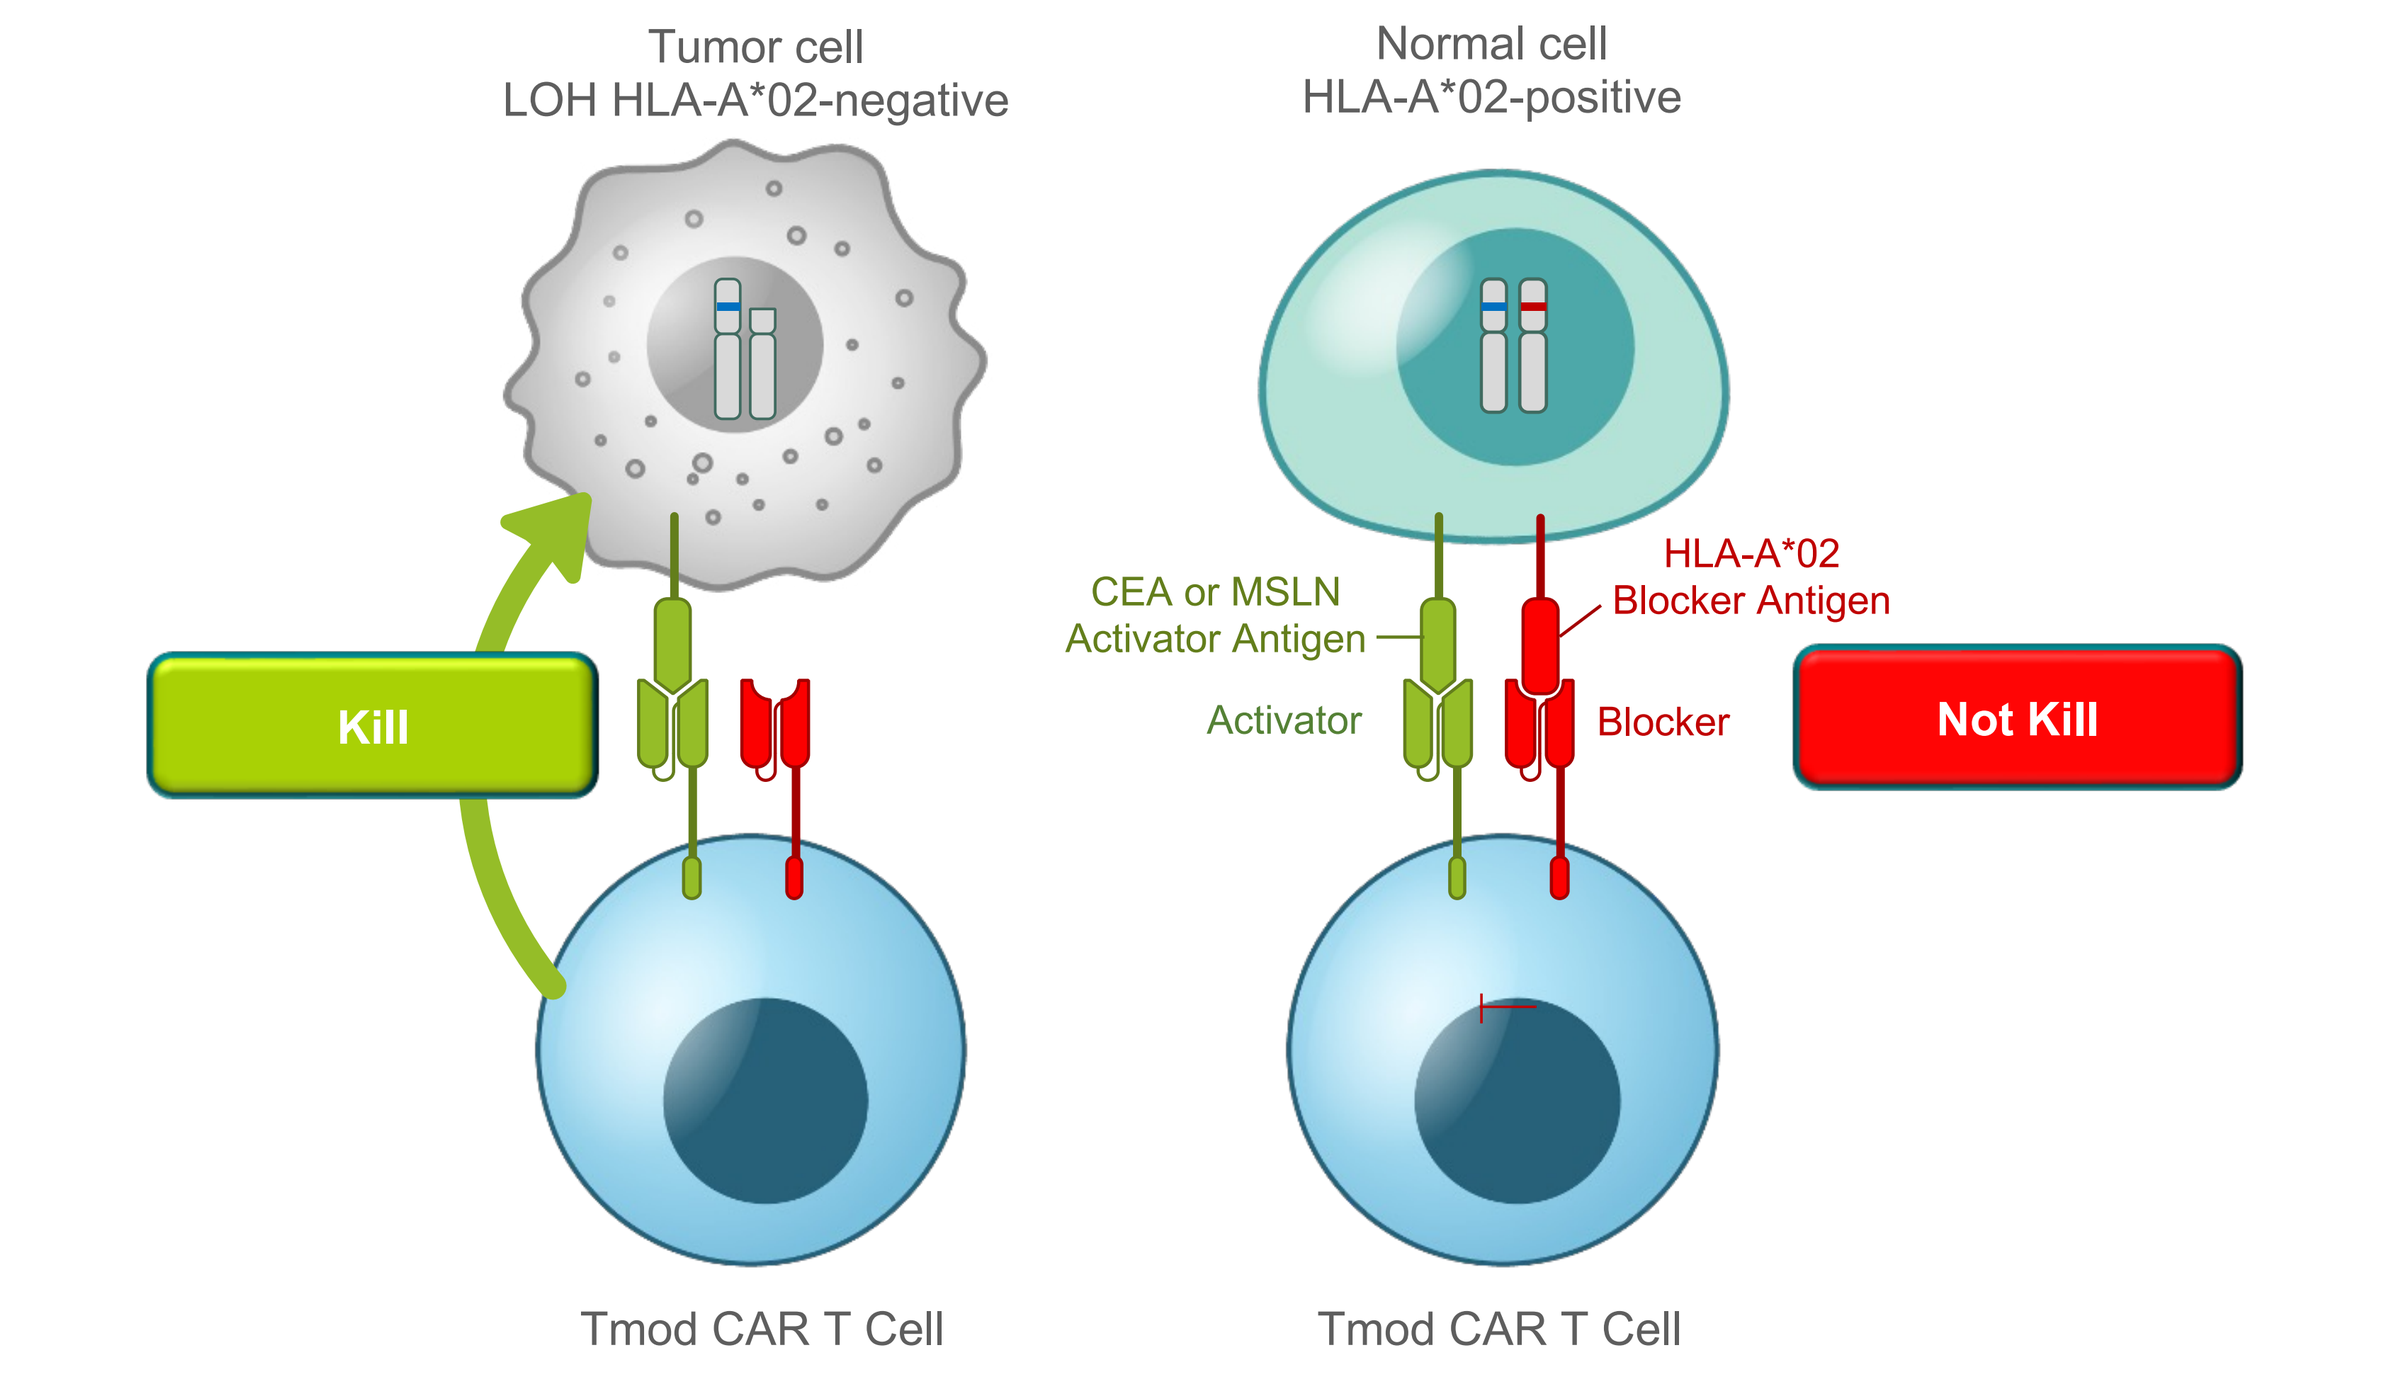
Tumor cell
LOH HLA-A*02-negative
Normal cell
HLA-A*02-positive
CEA or MSLN
Activator Antigen
HLA-A*02
Blocker Antigen
Activator
Blocker
Kill
Not Kill
Tmod CAR T Cell
Tmod CAR T Cell

CAR, chimeric antigen receptor; CEA, carcinoembryonic antigen; HLA, human leukocyte antigen; LOH, loss of heterozygosity; MSLN, mesothelin.

BACKGROUND (cont.)

Figure 2. CEA or MSLN CAR Tmod single vector construct [6]

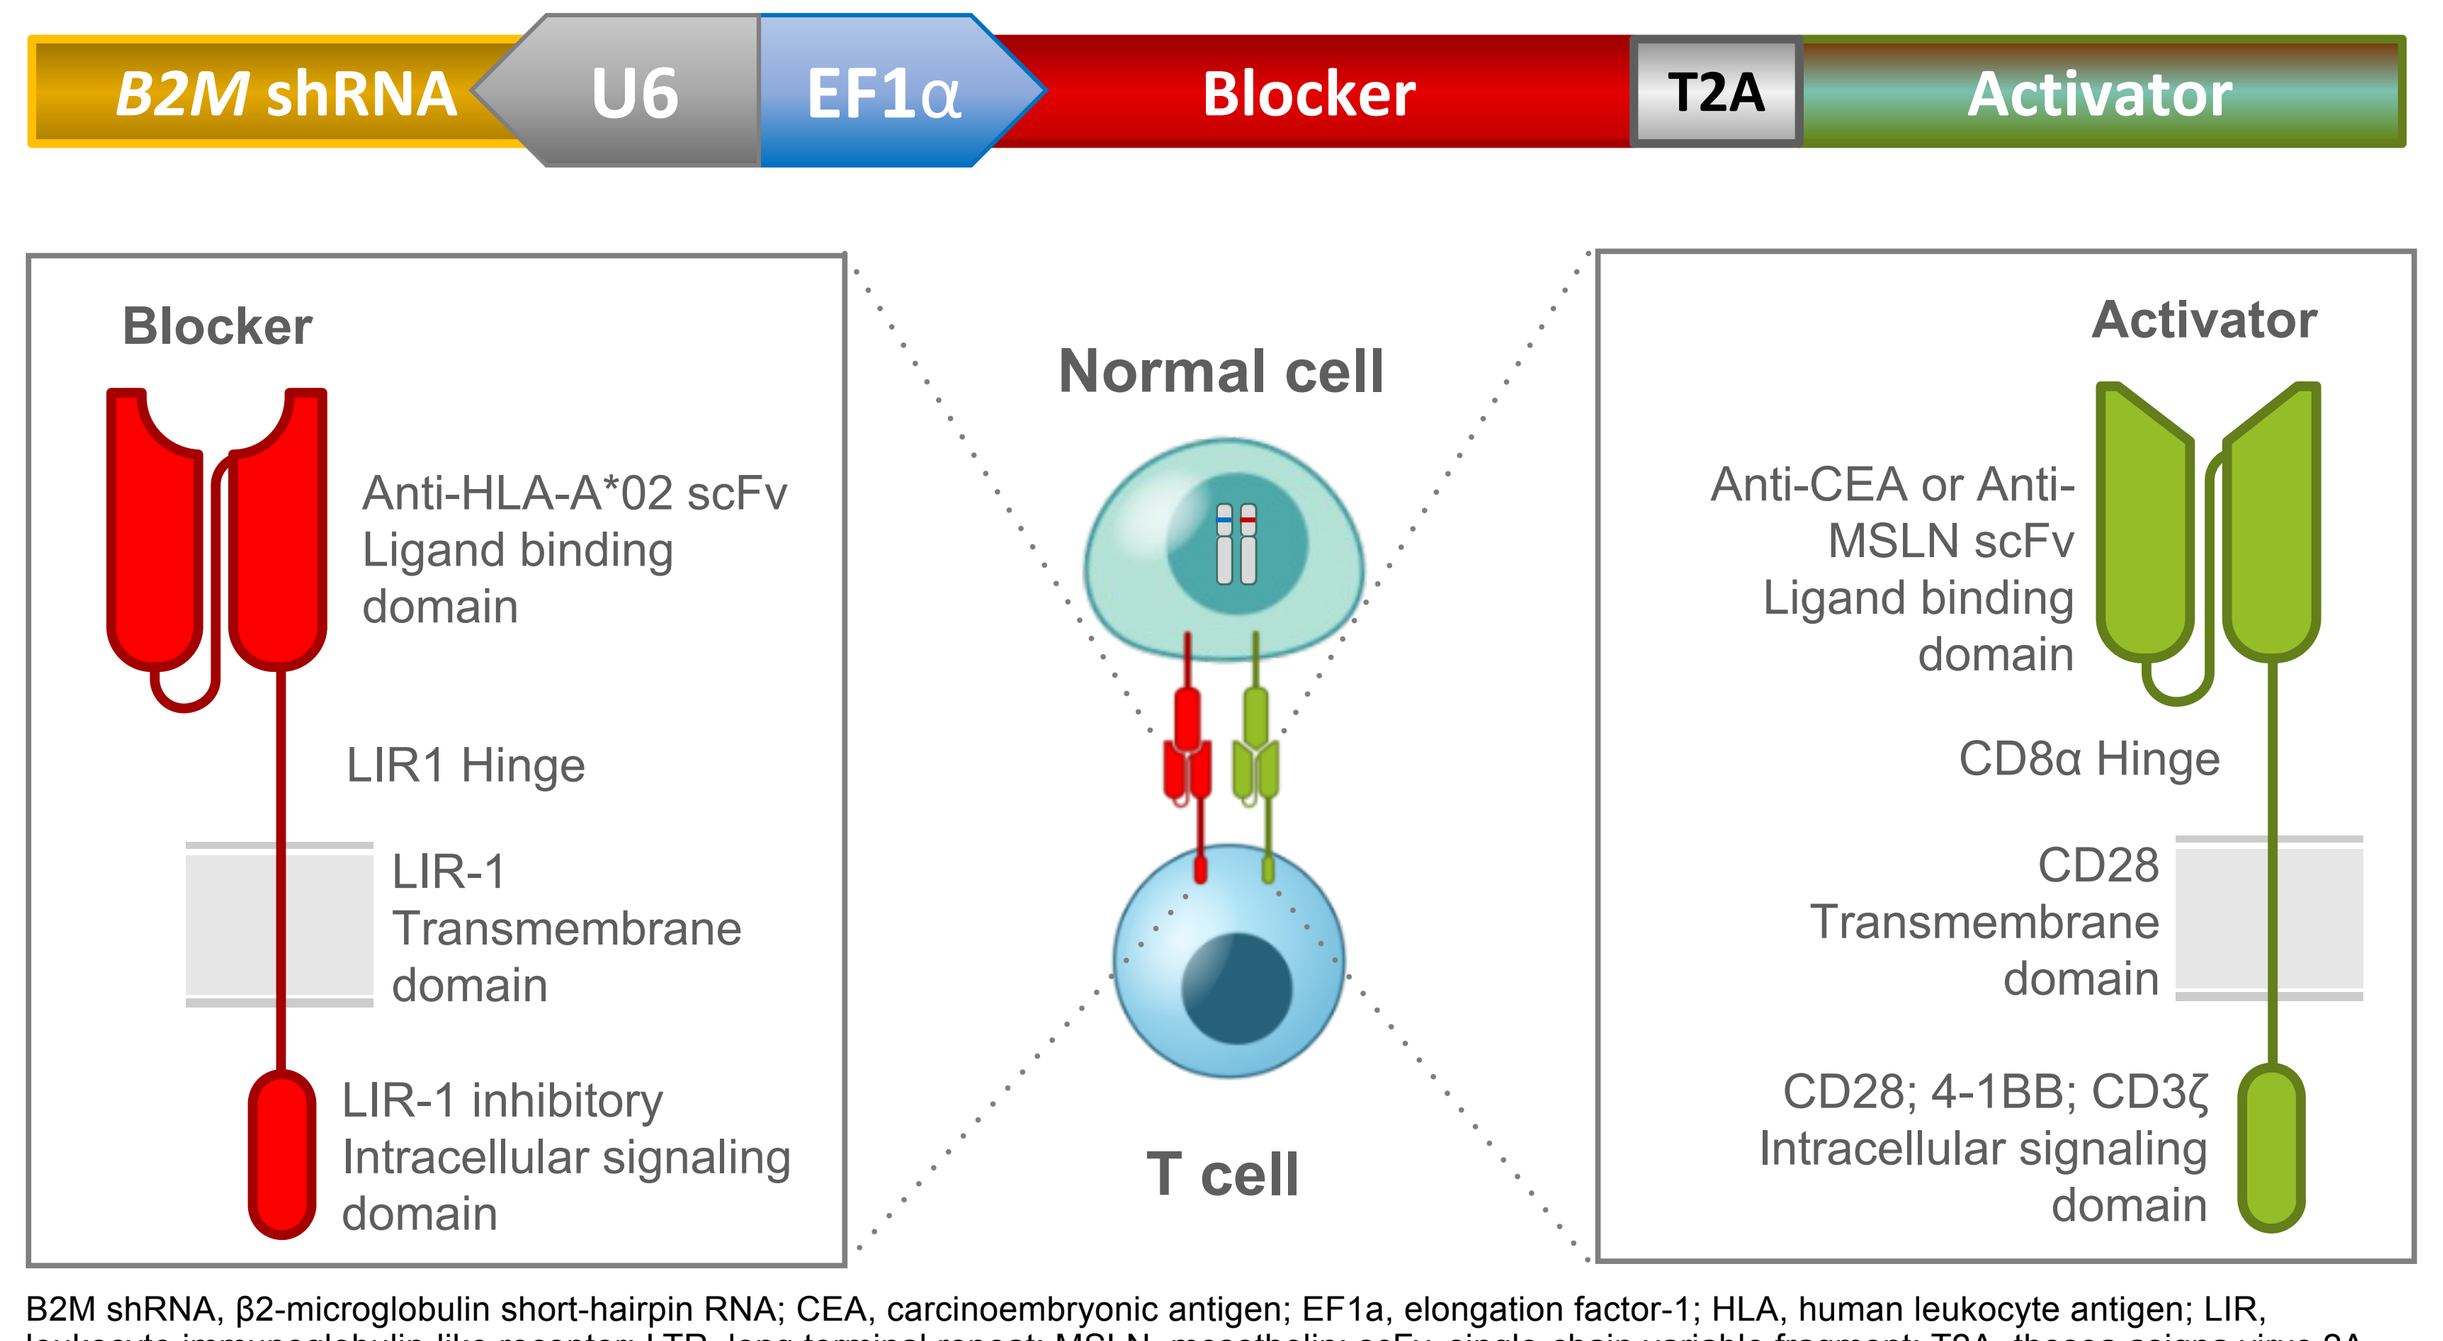
B2M shRNA
U6
EF1α
Blocker
T2A
Activator
Blocker
Anti-HLA-A*02 scFv
Ligand binding
domain
LIR1 Hinge
LIR-1
Transmembrane
domain
LIR-1 inhibitory
Intracellular signaling
domain
Normal cell
T cell
Activator
Anti-CEA or Anti-
MSLN scFv
Ligand binding
domain
CD8α Hinge
CD28
Transmembrane
domain
CD28; 4-1BB; CD3ζ
Intracellular signaling
domain

B2M shRNA, β2-microglobulin short-hairpin RNA; CEA, carcinoembryonic antigen; EF1a, elongation factor-1; HLA, human leukocyte antigen; LIR, leukocyte immunoglobulin-like receptor; LTR, long terminal repeat; MSLN, mesothelin; scFv, single-chain variable fragment; T2A, thosea asigna virus 2A

- CAR activator: 3rd-generation CAR T with both signal 1 (CD3ζ) and signal 2 activation domains (CD28 & 4-1BB)
- CAR blocker: Leukocyte immunoglobulin-like receptor 1 is a member of the immune inhibitory receptor family and contains 4 immunoreceptor tyrosine-based inhibition motifs in its signaling domain
- Replication-incompetent single lentivirus transgene: The activator and blocker receptors are co-expressed in a single construct containing a cleavable thosea asigna virus 2A linker (**Figure 2**)

METHODS

- De-identified records of patients with ≥stage 3 cancer were extracted from the Tempus Database and frequencies of HLA-A LOH were identified (ie, whether loss occurred across high-frequency HLA-A alleles)
- Using tumor/normal-matched DNA sequencing data from the Tempus xT assay, HLA-A LOH was determined by a research-use only device (**Figure 4**)
- Allele-specifc HLA LOH was identified in patients with additional resections or biopsies of tumors sent to Tempus as a standard-of-care diagnostic. Clonal loss of HLA-A*02 was determined based on exons 2 and 3 of HLA-A (**Figure 5**)
- Results were compared with previously published real-world datasets and stratified by other clinically relevant genetic features in colorectal cancer

Figure 3. HLA class I complex

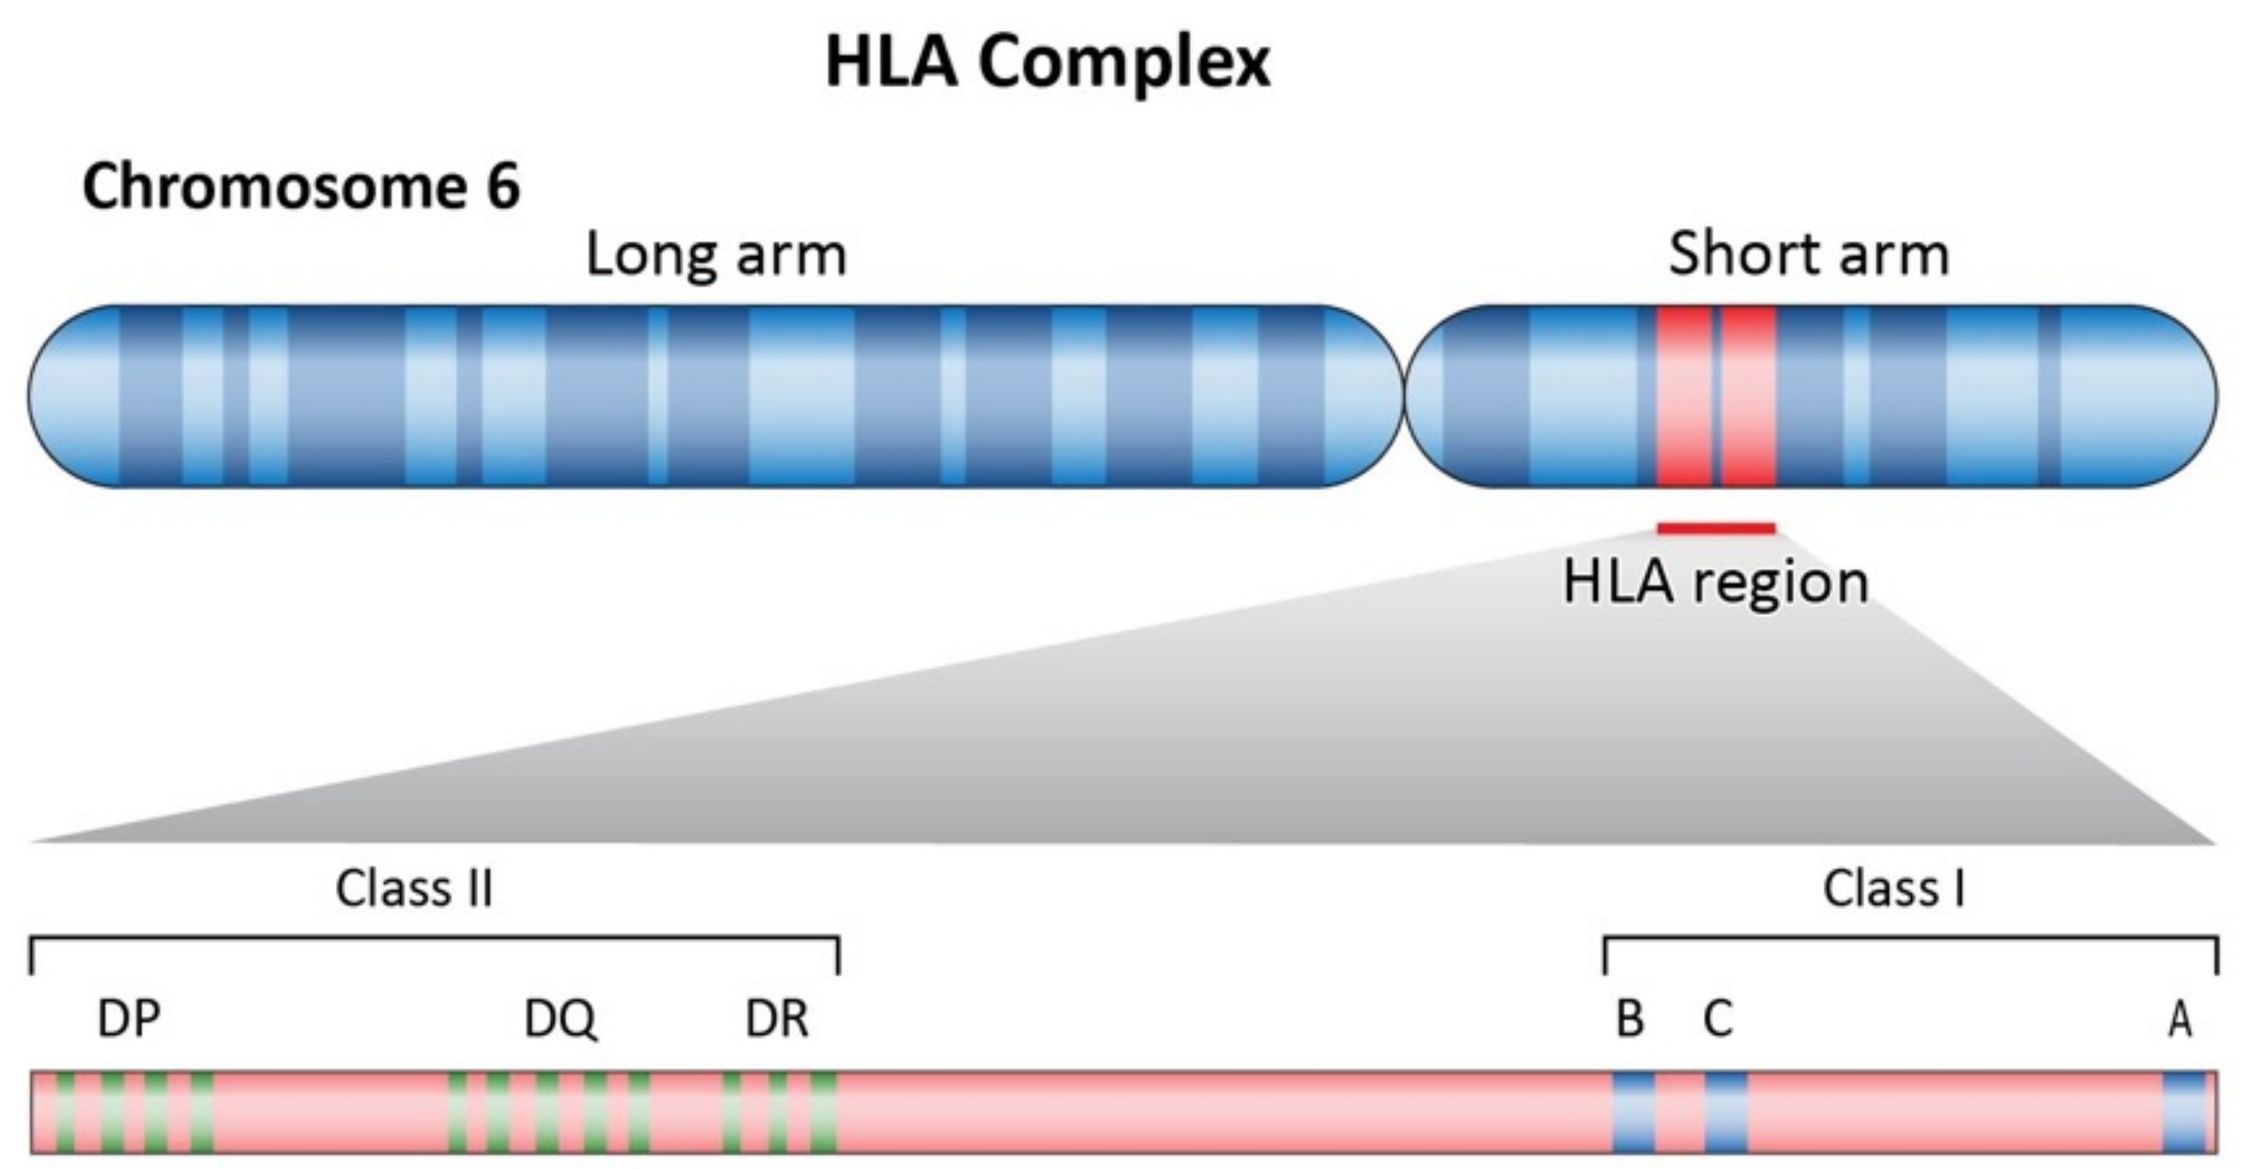
HLA Complex
Chromosome 6
Long arm
Short arm
HLA region
Class II
Class I
DP
DQ
DR
B
C
A

HLA, human leukocyte antigen.

METHODS (cont.)

Figure 4. Tempus HLA typing and analysis of HLA-A LOH

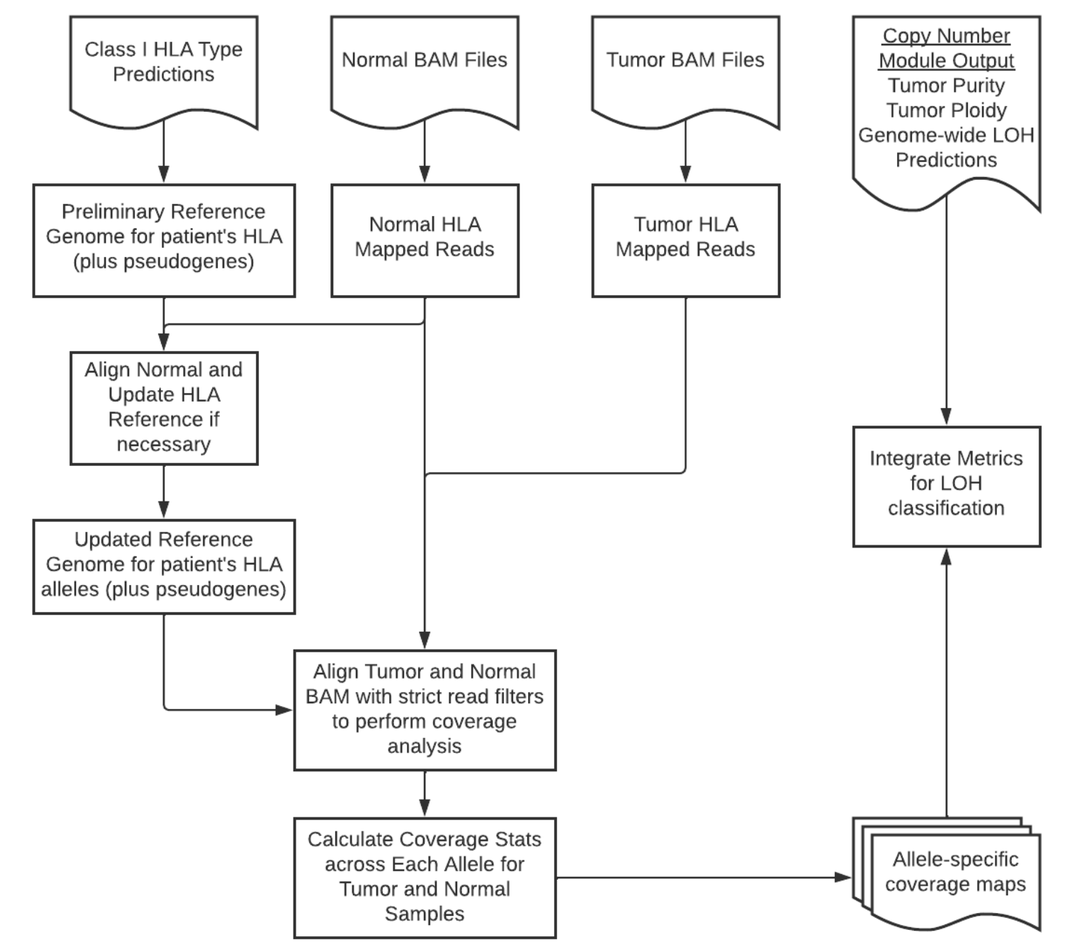
Class I HLA Type Predictions
Normal BAM Files
Tumor BAM Files
Copy Number Module Output
Tumor Purity
Tumor Ploidy
Genome-wide LOH Predictions
Preliminary Reference Genome for patient's HLA (plus pseudogenes)
Normal HLA Mapped Reads
Tumor HLA Mapped Reads
Align Normal and Update HLA Reference if necessary
Integrate Metrics for LOH classification
Updated Reference Genome for patient's HLA alleles (plus pseudogenes)
Align Tumor and Normal BAM with strict read filters to perform coverage analysis
Calculate Coverage Stats across Each Allele for Tumor and Normal Samples
Allele-specific coverage maps

BAM, Binary Alignment Mapping; HLA, human leukocyte antigen; LOH, loss of heterozygosity.

Ratio of allele 1 and allele 2

- To enhance our ability to understand differences in coverage, we can calculate higher-order coverage metrics
- In this case, the B allele frequency is the ratio of coverage for allele 1 and allele 2 at each position
- In a stable sample, the frequency for the two alleles should remain fairly close to 0.5; however, in HLA-A LOH, the frequencies will diverge

Figure 5. HLA-A LOH feature overview: B allele frequency

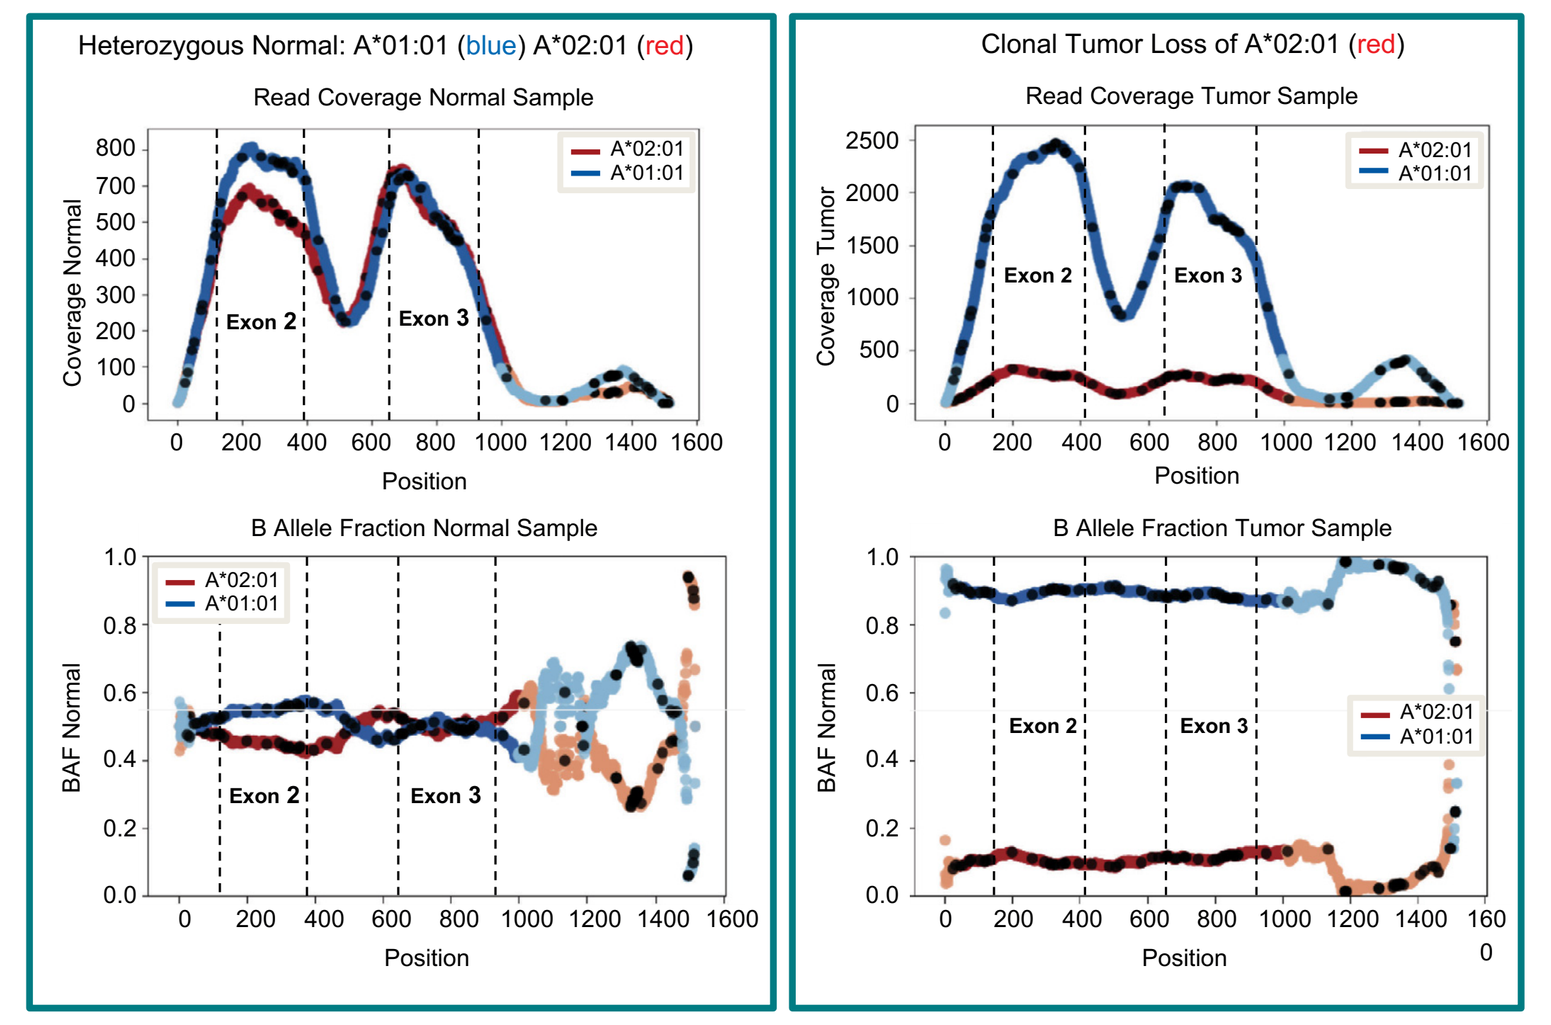
Heterozygous Normal: A*01:01 (blue) A*02:01 (red)
Read Coverage Normal Sample
Coverage Normal
Exon 2
Exon 3
Position
B Allele Fraction Normal Sample
BAF Normal
Clonal Tumor Loss of A*02:01 (red)
Read Coverage Tumor Sample
Coverage Tumor
B Allele Fraction Tumor Sample
A*02:01
A*01:01

HLA, human leukocyte antigen.

- Additional samples were assessed to determine the accuracy of the assay (**Figure 6**) using mixtures of negative and positive samples to determine the accuracy and sensitivity based on tumor purity

RESULTS

HLA-A LOH frequency across tumor types

- We applied bioinformatic HLA-A LOH detection across 11,828 samples previously sequenced at Tempus using the xT assay, representing real-world samples from 9 different tumor types
- We compared results with data reported from The Cancer Genome Atlas (TCGA) samples and from clinical samples evaluated within a second dataset of real-world patients [8,9]
- We observed HLA-A LOH in 1,841 patients with a median frequency of 18.8% across these 9 tumor types. Our observed tumor-specific HLA-A LOH frequency was very similar to previous data, with the exception of the modestly lower frequencies observed in prostate cancer: 3.3% of patients vs 5.8%
- Interestingly, we, like others [9], observed lower frequencies of HLA-A LOH in pancreatic vs TCGA data ($P<0.01$), but higher frequencies in colorectal and head and neck squamous samples (both $P<0.01$)
 - The TCGA dataset contained more primary and localized tumors, whereas our dataset contained more advanced disease, and this may have contributed to the higher proportion of patients we observed with HLA-A LOH
 - This also may reflect differences in patient selection in TCGA vs. real-world clinical diagnostic practice as well as statistical variance within the smaller TCGA sample set

RESULTS (cont.)

Figure 6. Sensitivity and specificity of HLA-A LOH detection

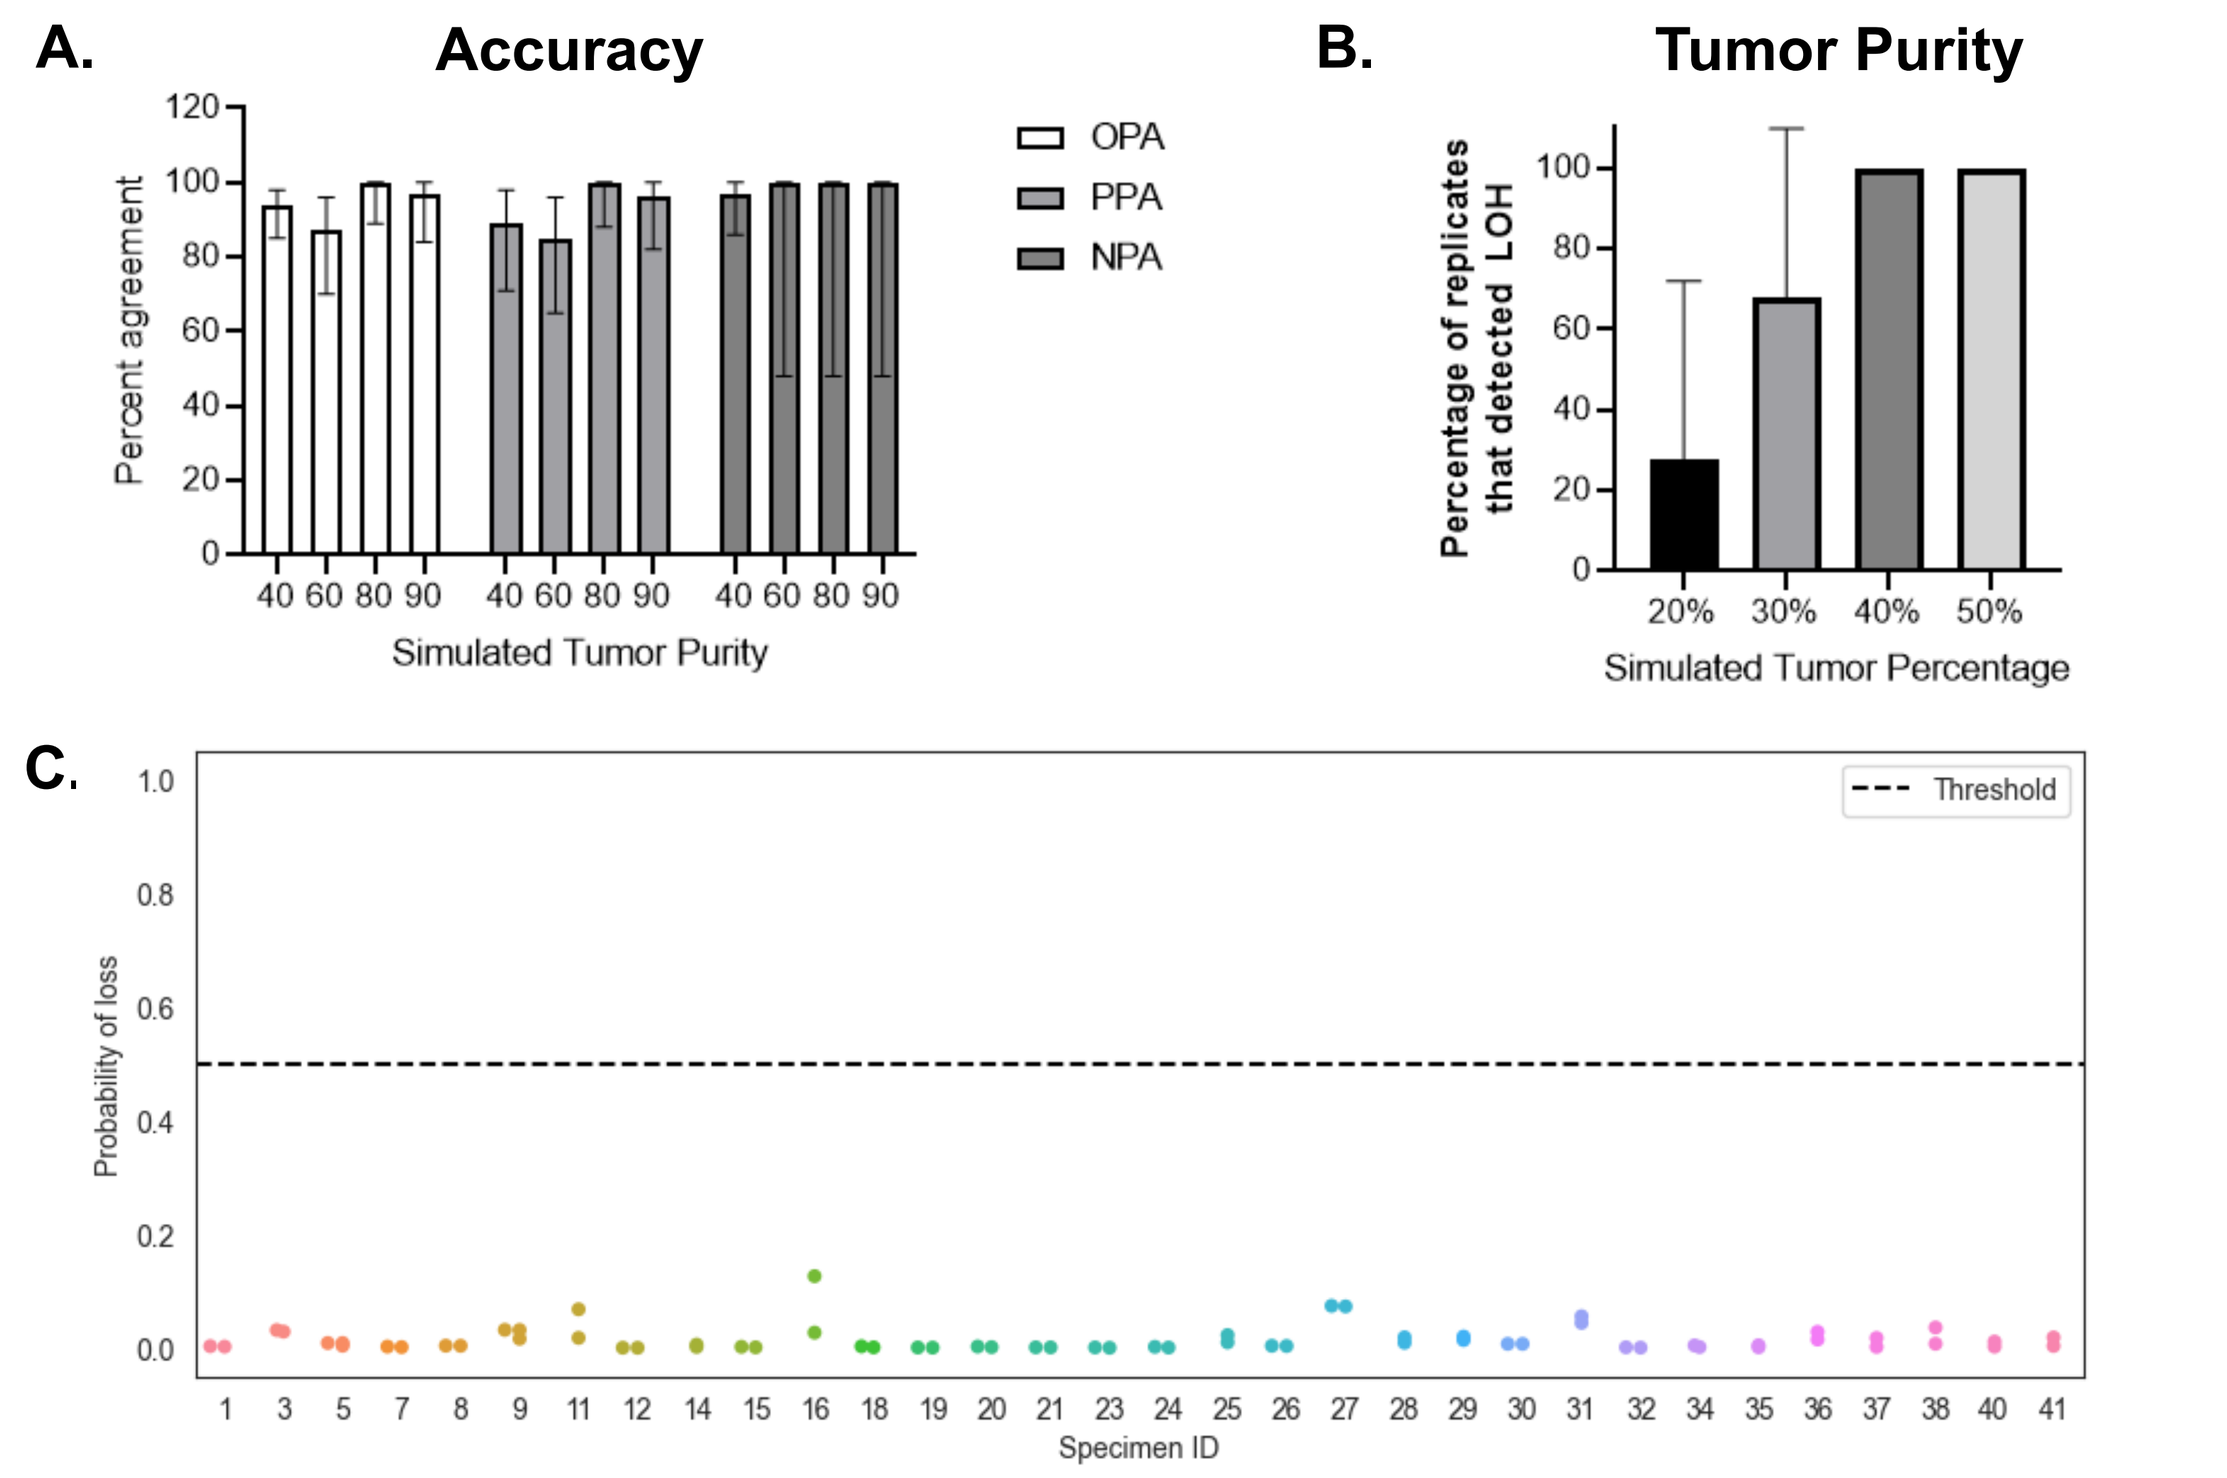
A.
Accuracy
Percent agreement
Simulated Tumor Purity
OPA
PPA
NPA
B.
Tumor Purity
Percentage of replicates that detected LOH
Simulated Tumor Percentage
C.
Probability of loss
Threshold
Specimen ID

A-B. A total of 162 samples were assessed for accuracy. DNA was generated for analysis from well-characterized cell lines and represented 17 different HLA-A alleles (A*01:01, A*02:05, A*03:01, A*11:01, A*23:01, A*24:02, A*25:01, A*26:01, A*29:01, A*29:02, A*30:01, A*30:02, A*31:01, A*32:01, A*33:01, A*66:01, A*68:02). DNA from two cell lines were combined to simulate three allelic categories: HLA-A*02:01 LOH, LOH in a non-HLA-A*02:01 allele, and no HLA LOH. **C.** A total of 32 clinical tumor samples with a native tumor purity of 20% along with the tumor-matched normal were run in replicates of 2 and processed through sequencing. Samples were selected from patients with colorectal cancer, non-small cell lung cancer, and pancreas cancer. All 32 clinical tumor samples were confirmed to be copy number stable by data generated from the Infinium CytoSNP-850K BeadChip Array. Thirteen HLA alleles were included (A*01:01, A*01:02, A*03:01, A*11:01, A*23:01, A*24:02, A*25:01, A*26:01, A*30:01, A*31:01, A*32:01, A*33:03, and A*68:01). All 32 samples were called negative.
HLA, human leukocyte antigen; LOH, loss of heterozygosity; NPA, negative percent agreement; OPA, overall percent agreement; PPA, positive percent agreement.

Table 2. Comparison of HLA-A LOH frequencies in three different datasets[a]

	Tempus [8]		TCGA [9]		Montesion et al [10]	
	Samples, n	**HLA-A LOH frequency, %**	**Samples, n**	**HLA-A LOH frequency, %**	**Samples, n**	**HLA-A LOH frequency, %**
Colorectal cancer	1854	15.6	615	9.6	10,682	15.3
Gastroesophageal cancer	506	20.8	625	16.2	3,174	22.2
Pancreatic cancer	675	19.6	184	33.1	4,049	23.4
Prostate cancer	998[b]	3.4[b]	500	4.5	2,774	5.8
Ovarian, fallopian tube, primary peritoneal cancers	569	16	579	17.1	4,996	15.7
NSCLC	1,915	23.1	501	25.3	13,240	23.0
Breast cancer	1447	12.2	1,080	13.6	9,686	13.2
Head and neck squamous cell carcinoma	208	26.0	522	16.1	1,134	27.2
Mesothelioma	7	14.3	87	11.5	404	12.4

[a] Tempus data contain more advanced disease, and TCGA data have more primary tumors; [b] Unpublished data.
HLA, human leukocyte antigen; LOH, loss of heterozygosity; NSCLC, non-small cell lung cancer; TCGA, The Cancer Genome Atlas.

Figure 7. HLA-A LOH is similar across multiple haplotypes

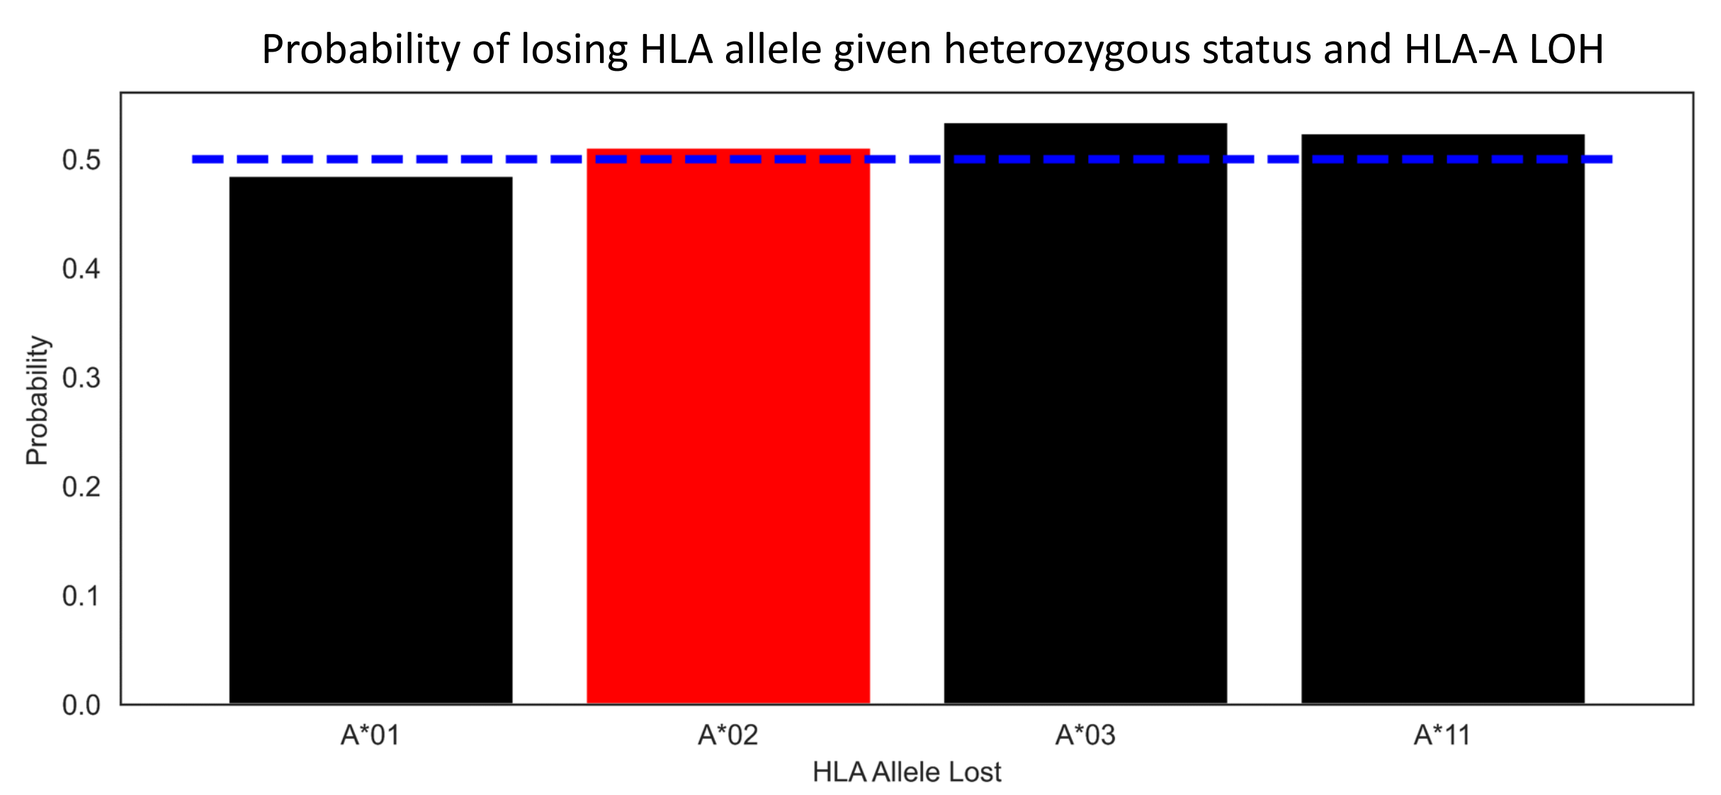
Probability of losing HLA allele given heterozygous status and HLA-A LOH
Probability
A*01
A*02
A*03
A*11
HLA Allele Lost

HLA, human leukocyte antigen; LOH, loss of heterozygosity.

RESULTS (cont.)

Figure 8. Major mutation frequencies are similar in patients with colorectal cancer with and without HLA-A LOH

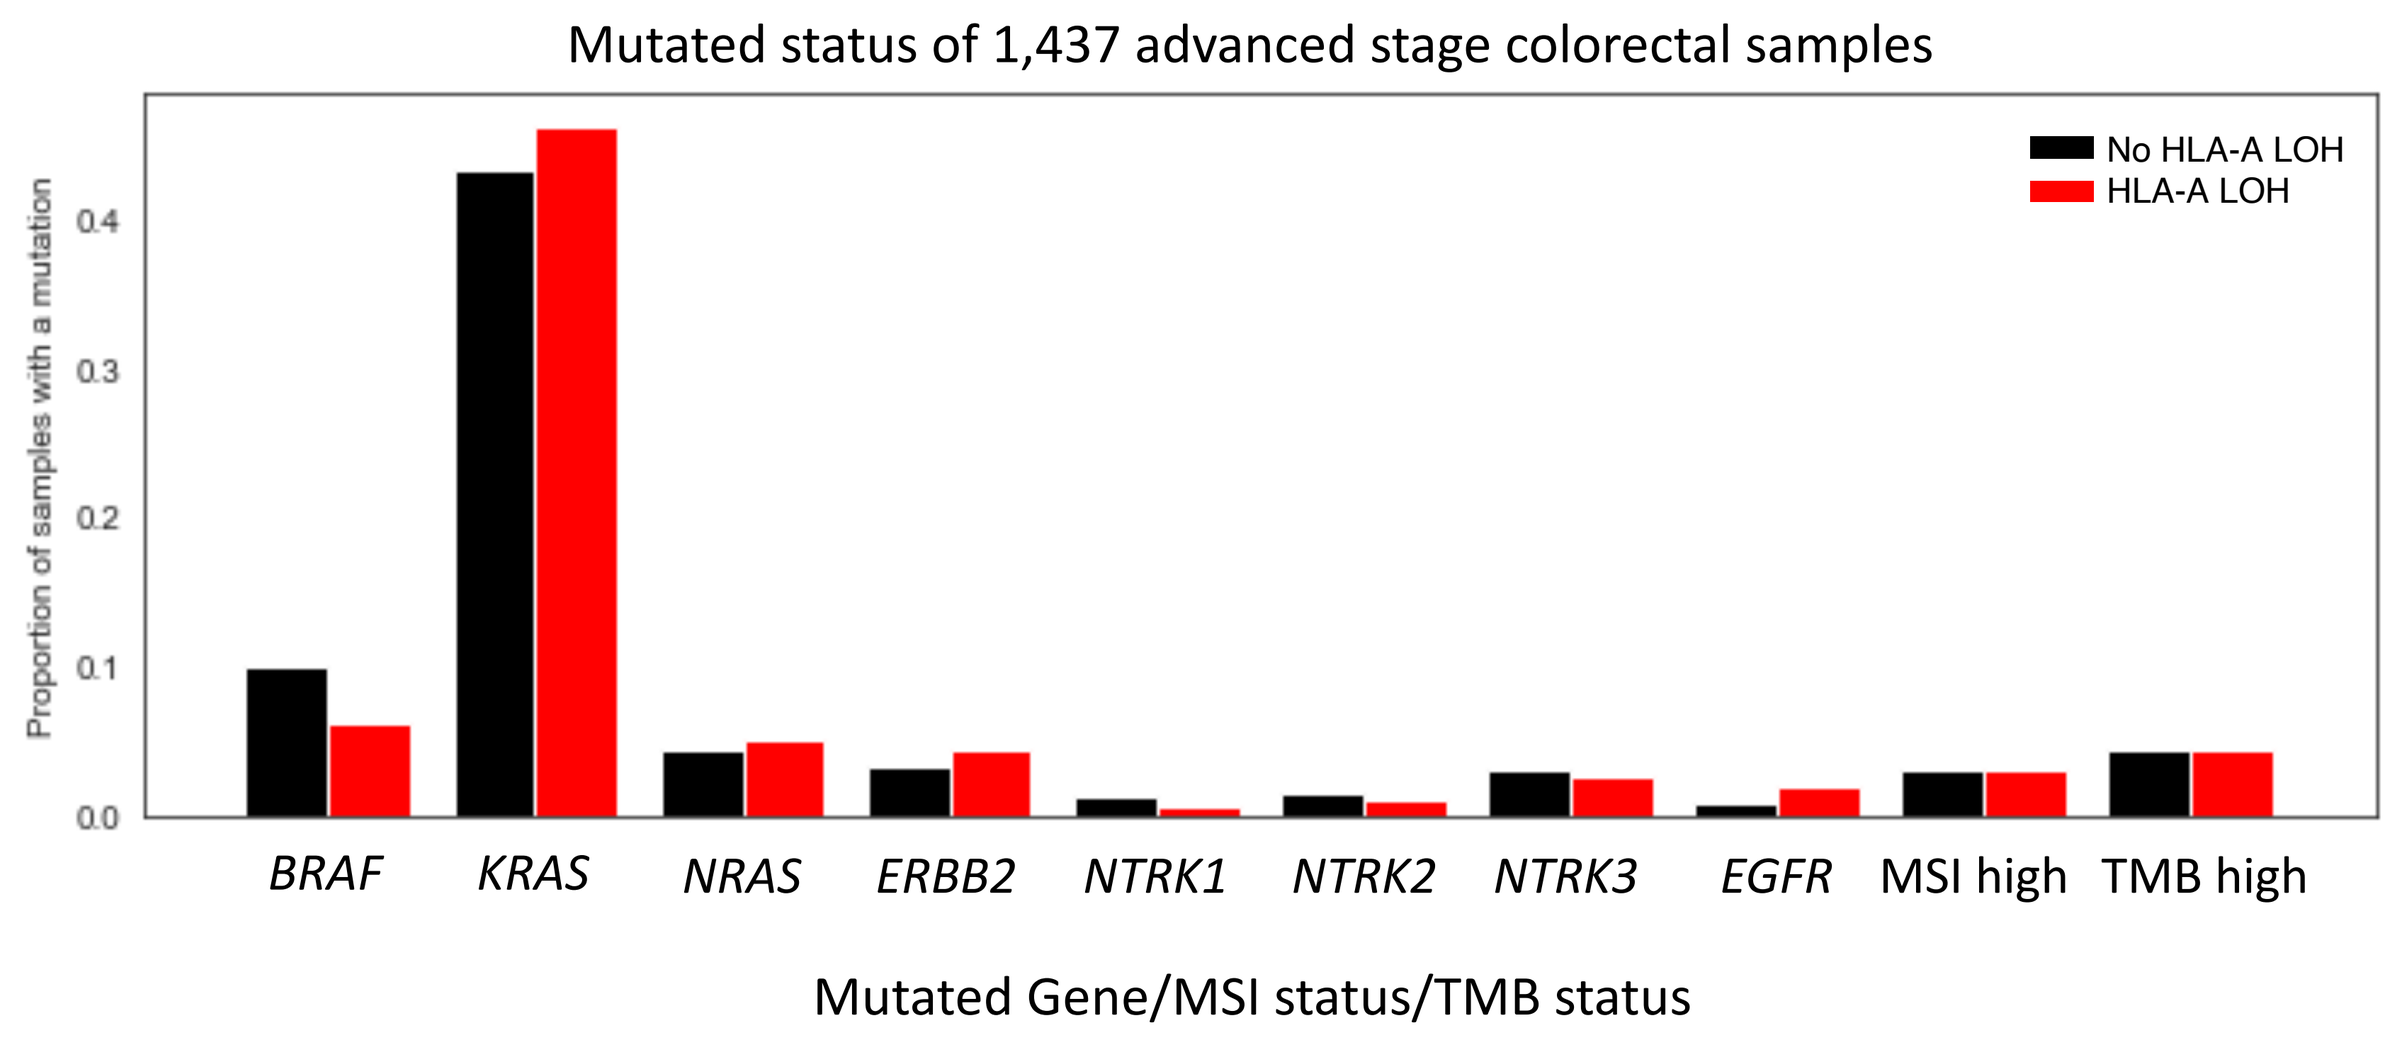
Mutated status of 1,437 advanced stage colorectal samples
No HLA-A LOH
HLA-A LOH
Proportion of samples with a mutation
BRAF
KRAS
NRAS
ERBB2
NTRK1
NTRK2
NTRK3
EGFR
MSI high
TMB high
Mutated Gene/MSI status/TMB status

BRAF, v-Raf murine sarcoma viral oncogene homolog B; *EGFR*, epidermal growth factor receptor; HLA, human leukocyte antigen; *KRAS*, v-Ki-ras2 Kirsten rat sarcoma viral oncogene homolog; LOH, loss of heterozygosity; MSI, microsatellite instability; *NRAS*, neuroblastoma rat sarcoma; *NTRK1/2/3*, neurotrophic tyrosine receptor kinase 1/2/3; TMB, tumor mutation burden.

CONCLUSIONS

- Tempus xT standard-of-care NGS has the ability to identify real-word patients with clonal HLA-A LOH
- Clonal HLA-A LOH is a clear foundational distinguisher between tumor and normal cells [6,7]
- The frequency of HLA-A LOH among advanced solid tumor cancers in this dataset is 15.5% with variable frequency across tumor types
- The HLA-A LOH frequency observed across diverse solid tumors is consistent with results from TCGA and samples evaluated at Foundation Medicine in an additional dataset [9,10]
- Tempus NGS was able to identify real-world patients with clonal HLA-A LOH, which can be used for Tmod CAR-T cell therapy to an enhanced therapeutic window
- BASECAMP-1 (NCT04981119) will identify and enroll HLA-A LOH patients with GI malignancies to bank their own T cells for future EVEREST novel Tmod CAR T-cell therapy interventional trials

References

1. American Cancer Society. Cancer Facts & Figures 2022. Atlanta: American Cancer Society; 2022.
2. Locke F, et al. *N Engl J Med.* 2022;386(7):640-654.
3. Maude S, et al. *N Engl J Med.* 2018;378(5):439-448.
4. Parkhurst M, et al. *Mol Ther.* 2011;19(3):620-626.
5. Haas AR, et al. *Mol Ther.* 2019;27(11):1919-1929.
6. Hamburger A, et al. *Mol Immunol.* 2020;128:298-310.
7. Hwang M, et al. *Proc Natl Acad Sci U S A.* 2021;118(12):e2022410118.
8. Hecht J, et al. J Clin Oncol. 2022; 40(4_suppl):190-190.
9. The Cancer Genome Atlas (TCGA) Research Network. Accessed June 2021. https://www.cancer.gov/tcga
10. Montesion M, et al. *Cancer Discov.* 2021;11(2):282-292.

Acknowledgments

The authors would like to thank:
- Alexander Kamb, PhD – Founder and Chief Scientific Officer of A2 Bio
- Han Xu, PhD – Vice President of Therapeutic Technology at A2 Bio
- Agnes E. Hamburger, PhD – Vice President of Drug Discovery at A2 Bio
- Mark L. Sandberg, PhD – Scientific Director of Therapeutic Technology at A2 Bio
- Talar Tokatlian, PhD – Senior Scientist at A2 Bio
- Alain Silk, PhD – Director of Regulatory Affairs at Tempus
- Bio Connections, LLC

This study was supported by A2 Bio

Presented at the SITC 37th Annual Meeting, November 10-12, 2022.